Abstract # LB-345

GlobeImmune
www.GlobeImmune.com

ONCOGENIC SYNERGY AND A NEWLY IDENTIFIED TRANSFORMING RAS MUTATION IN HUMAN CANCER

Zhimin Guo[1], Sarah Oakes[1], Yingnian Lu[1], Michio Sugita[2], Wilbur Franklin[2] & Alex Franzusoff[1]
[1]GlobeImmune Inc., Louisville, CO; [2]Univ. Colorado Health Sciences Ctr (UCHSC) Aurora, CO

Abstract

Aberrant signaling through the Ras oncogene product pathway plays an important role in uncontrolled cell proliferation and tumorigenesis. The well-characterized mutations at codons 12, 13 and 61 cause constitutive Ras activation. In this study, K-, N- and H-*ras* DNA sequences were characterized for the presence of tumor-associated mutations by nested PCR amplification and direct sequencing from tumors of 149 subjects with pancreas (68% harboring mutations), colorectal (40% with mutations) or NSCLC (9% with mutations) cancers in a phase 1 immunotherapy trial of whole, heat-killed yeast expressing mutated Ras proteins (Tarmogens). A new *ras* mutation at codon 76 was detected in 24 subjects from all 3 cancer types, with 22 being E76G, while 1 tumor each harbored E76K or E76Q mutations. Double combinations of E76 plus mutations at codons 12 or 13 were identified in 8 tumors.

Ras E76G and E76K mutations were confirmed as transforming in non-clinical studies. Of particular interest, coupling codon 76 and 12 mutations resulted in tumor growth synergy. G12V, E76G and E76K single mutations or double G12V-E76G mutations were introduced into the mouse K-*ras* gene and then transfected into BALB/3T3 fibroblasts. Cells transfected with *ras* harboring G12V or E76K alone, or the G12V-E76G double mutation formed colonies in soft agar. The BALB/3T3 cells transfected with wild-type or mutant K-*ras* genes were injected s.c. into BALB/c nude mice. The double G12V-E76G Ras mutation led to accelerated tumor growth compared to cells bearing any single mutation.

Mutations at codon 12 or codon 61 block γ-phosphate release from GTP or prevent Ras-GAP protein binding to Ras to trigger GTP hydrolysis, respectively. From the Ras crystal structure, residues 61 and 76 are positioned at opposing ends of the Switch 2 region α-helix responsible for Ras-GAP binding, implying that codon 76 mutations could similarly interfere with normal Ras-GTPase activity, albeit possibly in a less pronounced fashion than codons 12, 13 or 61 mutations, which could explain why this locus may have escaped attention until now. However, these results suggest that tumors bearing double mutations are more likely to exhibit aggressive growth characteristics. Thus, the genotype of double codon 12 and 76 *ras* mutations in patient tumors may be prognostic of an accelerated malignant phenotype. The identification of codon 76 *ras* mutations in human tumors also represents a new target for cancer therapy.

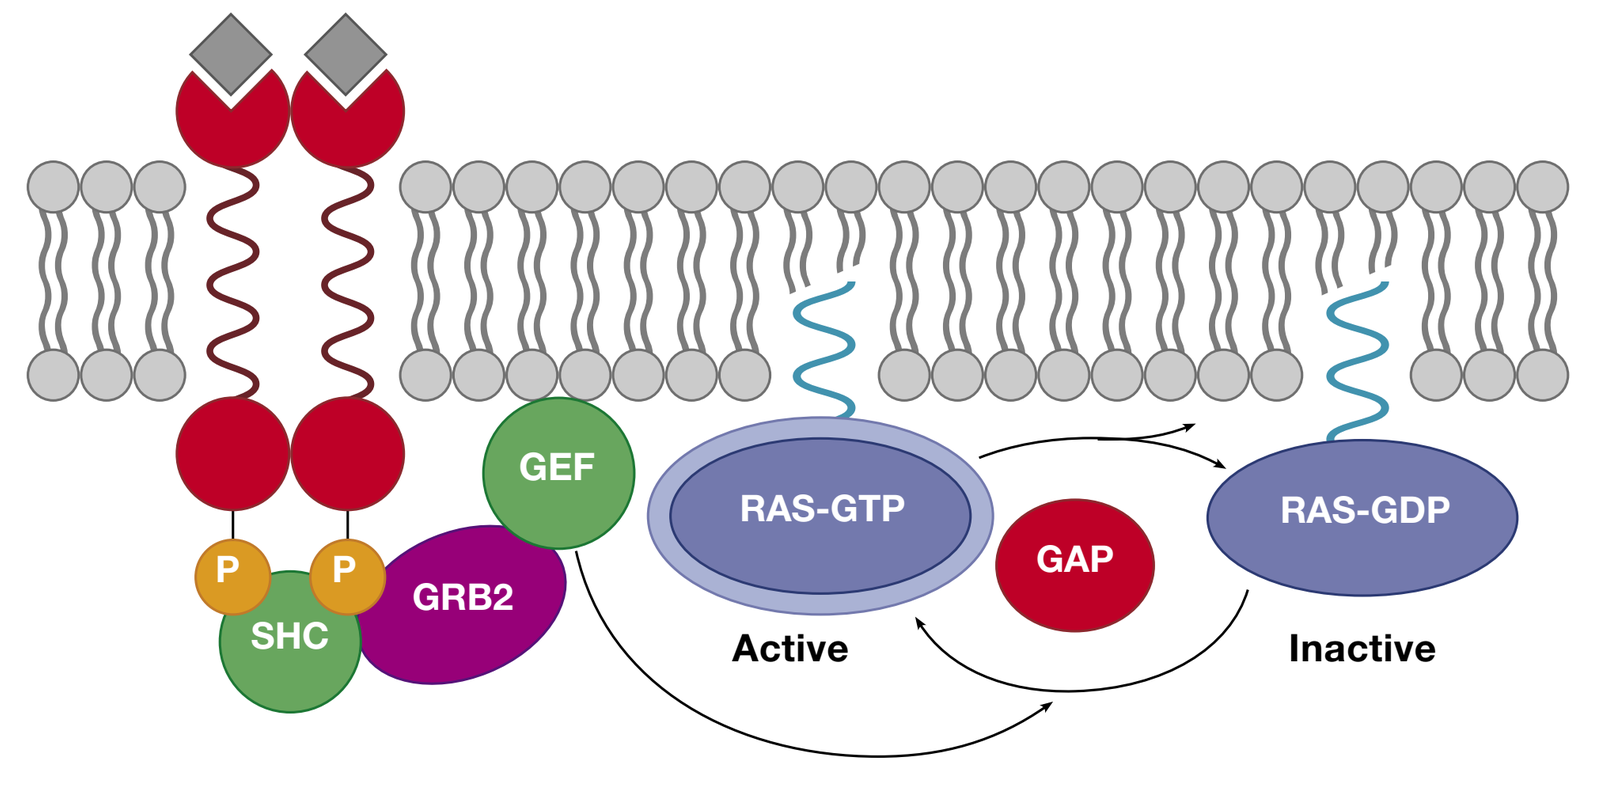
GEF
RAS-GTP
GAP
RAS-GDP
P
P
SHC
GRB2
Active
Inactive

Downward J. Nat Rev Cancer. 2003; 3 (1):11-22.

Ras and Ras binding proteins for signaling cell proliferation

The activation state of Ras is controlled by the cycle of hydrolysis of bound GTP, which is catalysed by GTPase activation proteins (GAPs), and the replacement of bound GDP with fresh GTP, which is catalysed by guanine nucleotide exchange factors (GEFs). In its active, GTP-bound state, Ras will interact with several families of effector proteins, resulting in stimulation of their catalytic activity.

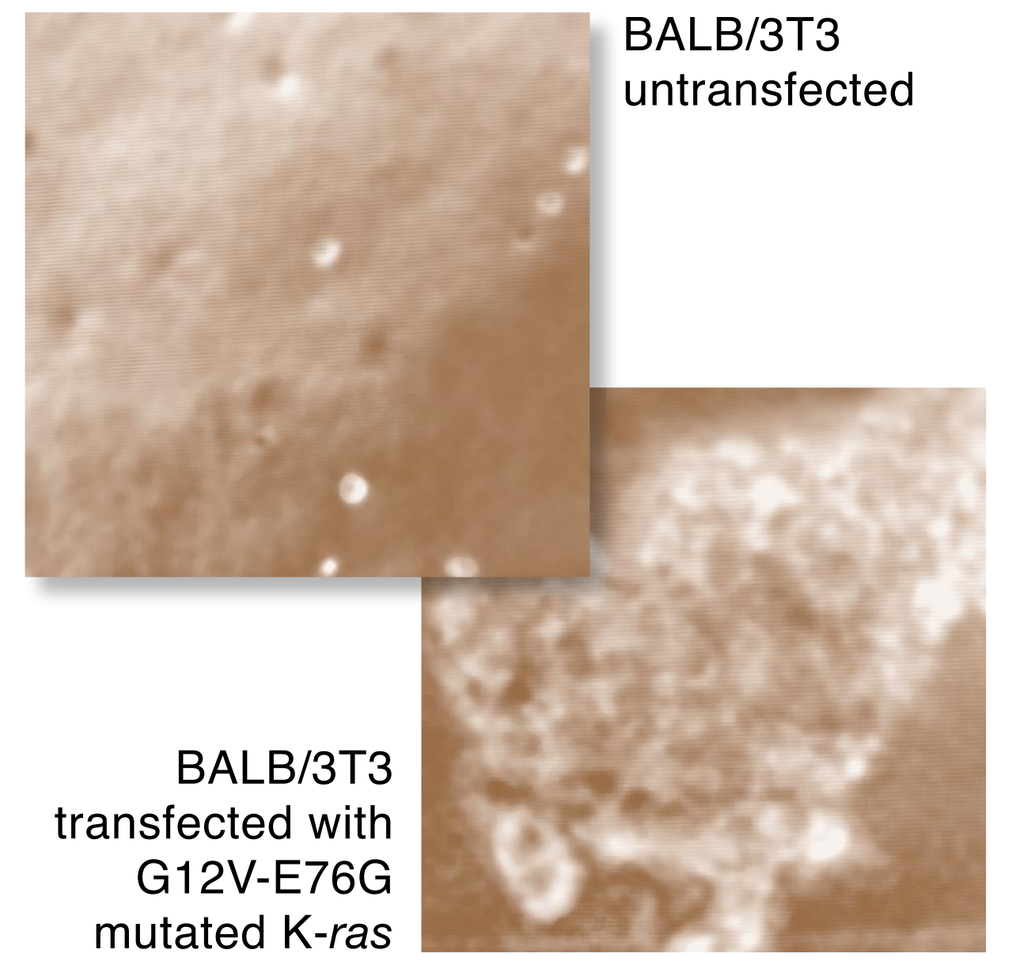
BALB/3T3
untransfected
BALB/3T3
transfected with
G12V-E76G
mutated K-*ras*

Figure 2 and Table 2 : Ras mutations cause transforming growth phenotypes in vitro

G12V, E76G and E76K single mutations or double G12V-E76G mutations were introduced into the mouse K-*ras* gene by site-directed mutagenesis and then transfected into BALB/3T3 fibroblasts. Cells transfected with *ras* harboring G12V or E76K alone, or the G12V-E76G double mutation (right panel) formed colonies in the soft agar growth assay, whereas untransformed cells (left panel) do not grow in soft agar.

Results

The *ras* gene family is one of the most frequently activated oncogenes in human cancer. Mutations at the codon encoding amino acid 12 in the Ras protein are reported in 78% pancreatic cancers, 34% colorectal cancers, 27% non-small cell lung carcinomas, and 24% ovarian cancers. Mutations at positions 13, 59 or 61 are also found in a variety of cancers. Codon 12 or 13 mutations affect γ-phosphate release from GTP, while mutations at codons 59 or 61 block Ras-GAP binding preventing GTP hydrolysis (see figure 4). Both classes of alterations result in constitutive Ras activation and uncontrolled cell proliferation.

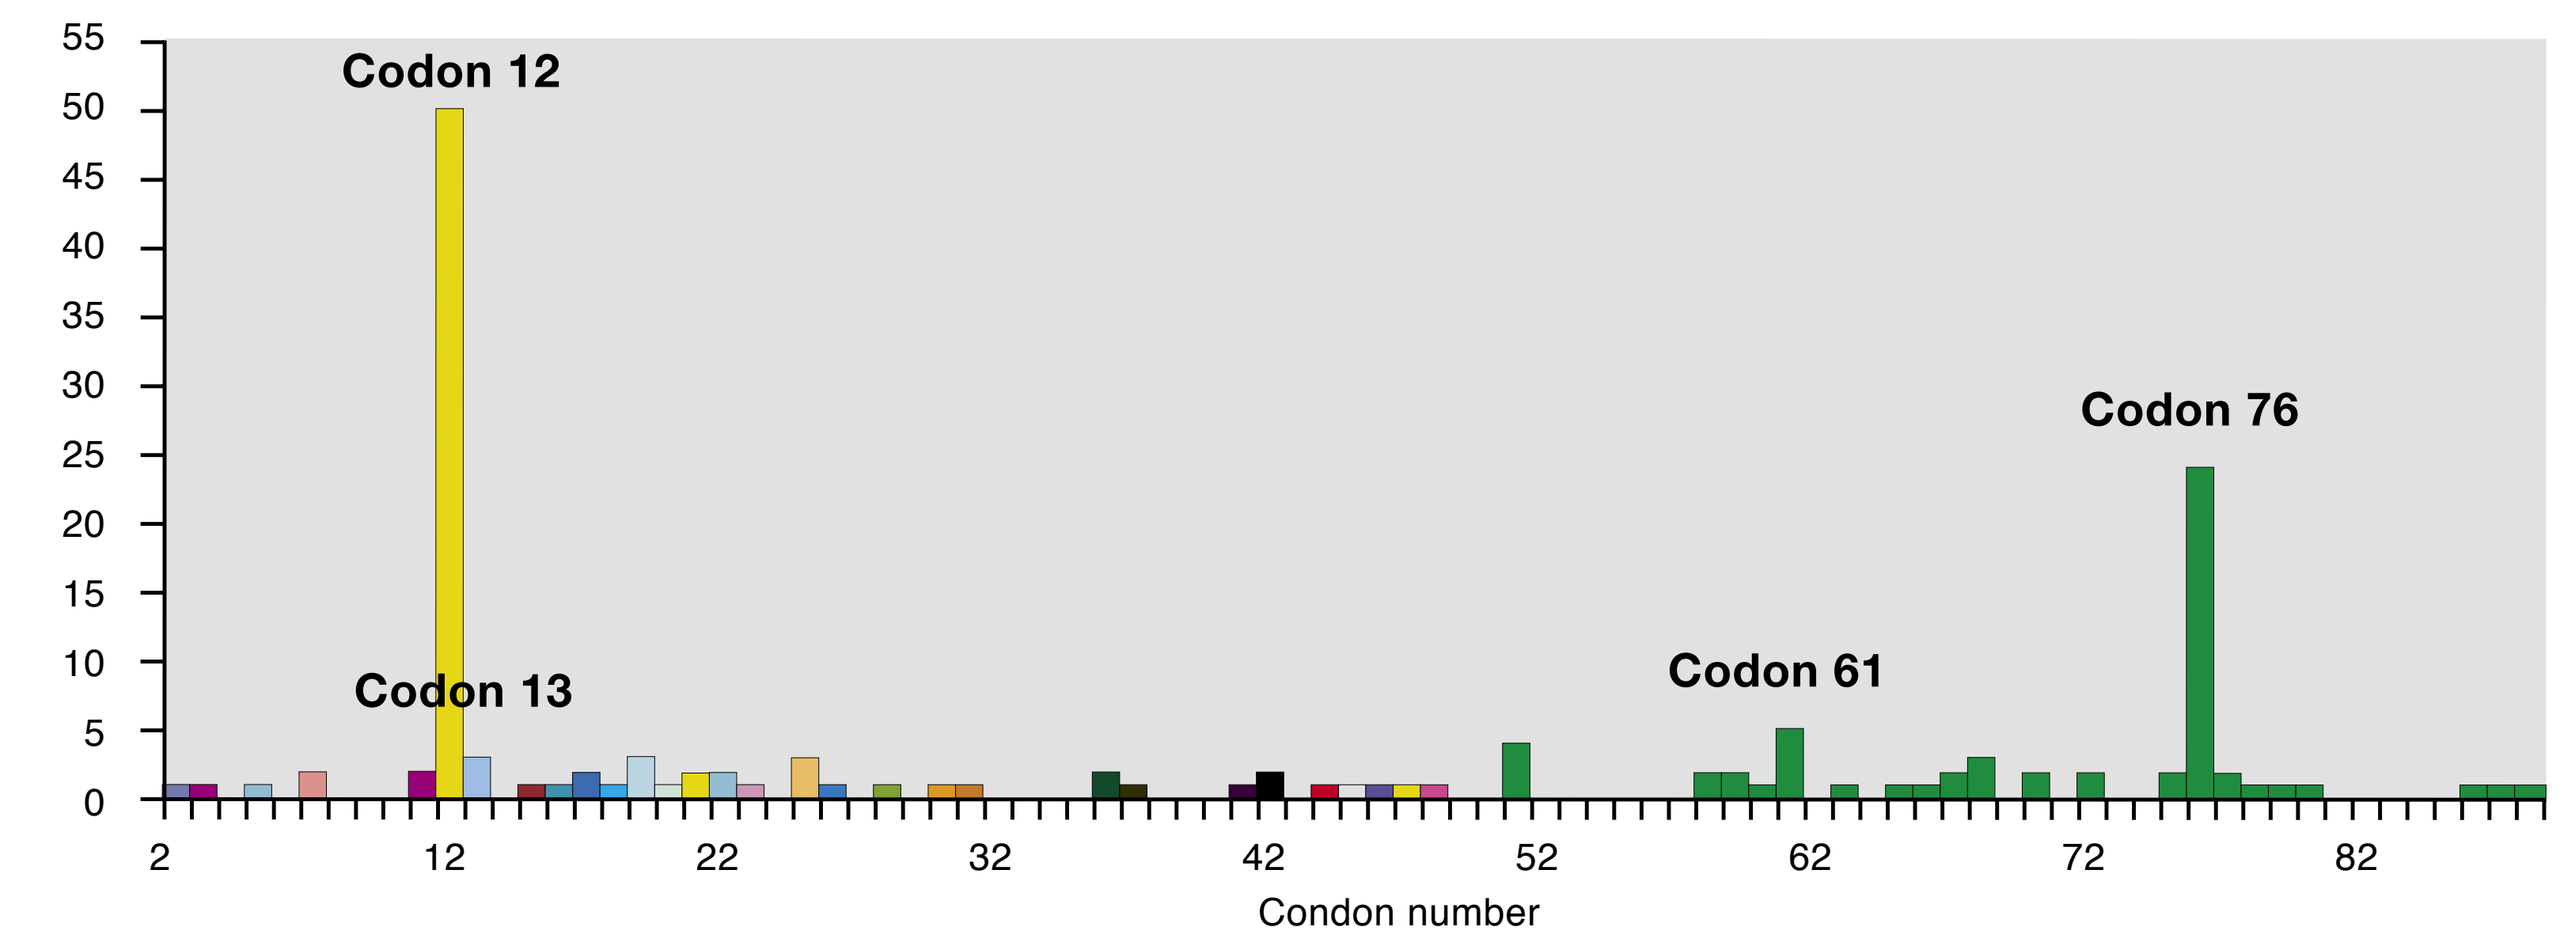
Codon 12
Codon 13
Codon 61
Codon 76
Condon number

Figure 1: Detection of *ras* mutations in phase I trial biopsy samples

Exons 2 and 3 of human K, N and H-*ras* genes from a total of 149 tumor samples (with pancreas, colorectal or NSCLC) were amplified by nested PCR and sequenced. Codon 12 mutations were found in 50 out of 149 samples, while codon 76 mutations was found in 24 out 149 samples. In addition, codon 13 mutations were found in 3 samples and codon 61 mutations were found in five samples.

	Colorectoral (85)			Lung (33)			Pancreas (31)			Total (149)
	K-ras	N-ras	H-ras	K-ras	N-ras	H-ras	K-ras	N-ras	H-ras	
E76G	2*		3*	3		1	2			
Possible E76G#	5		1			1	4		1	
E76K		1								
E76Q	1									
Total	8*	1	4*	3	0	2	6	0	1	24

* One sample has E76G in both K and H-*ras*;
\# weaker signal for E76 mutation detected, likely due to low abundance of tumor cells in biopsy sample.

Table 1: Summary of mutations at codon 76

A new *ras* mutation at codon 76 was detected in 24 subjects from all 3 cancer types, with 22 being E76G mutations, while 1 tumor harbored an E76K and 1 tumor had an E76Q mutations. Double combinations of E76 plus mutations at codons 12 or 13 were identified in 8 tumors.

Clone (K-*ras* genotype transfected into BALB/3T3 cells)	mRNA transcript of transfected *ras* gene (by RT-PCR)	Exogenous Ras protein expression (western blot)	Colony formation in soft agar
wild-type (WT) K-*ras*	+	+	-
G12V	+	+	++++
G12V + E76G	+	+	+++++
E76G	+	+	-
E76K	+	+	++
pUP (empty vector)	-	-	-
untransfected	-	-	-

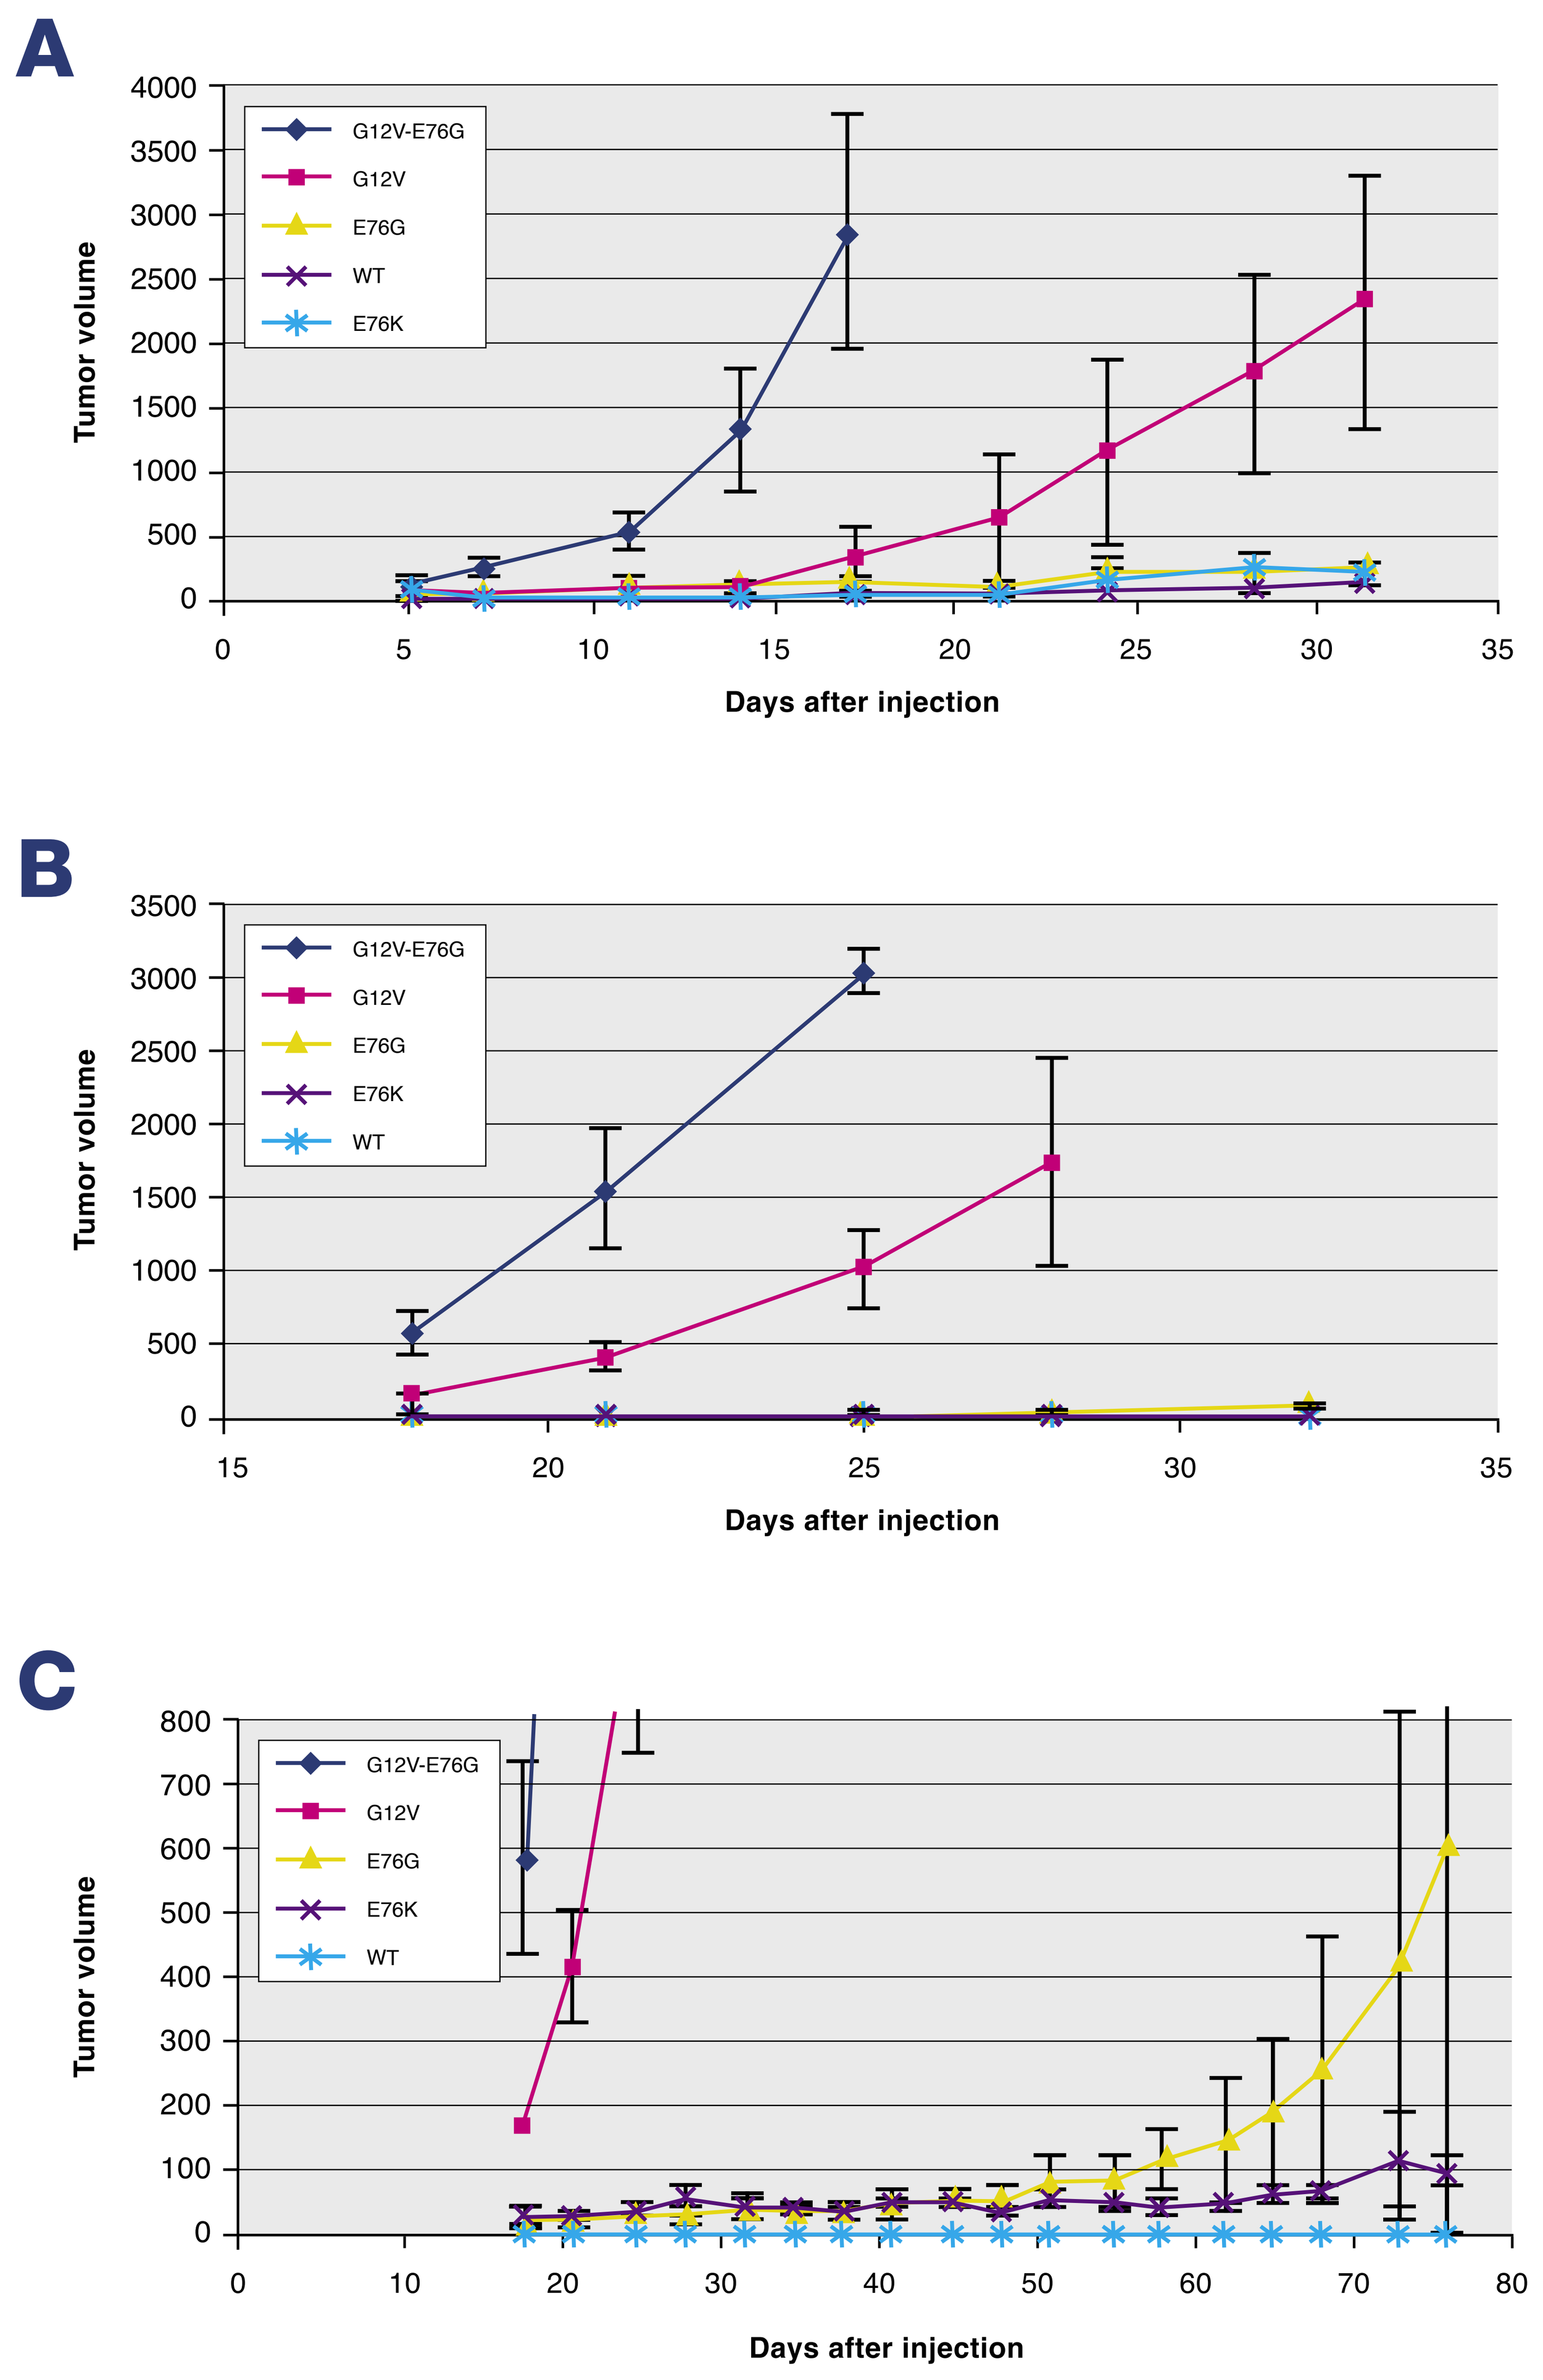
A
Tumor volume
Days after injection
G12V-E76G
G12V
E76G
WT
E76K
B
Tumor volume
Days after injection
G12V-E76G
G12V
E76G
E76K
WT
C
Tumor volume
Days after injection
G12V-E76G
G12V
E76G
E76K
WT

Figure 3: Tumorigenicity assay with Balb/c nude mice

Each transfected cell line was grown, harvested, and suspended in PBS. Five million cells were injected s.c. into 4-6-week-old Balb/c nude mice. Tumors were measured and tumor volume was calculated as 3.14 x (length) x (width)^2 / 6. Figs. 3A and 3B show two separate experiments demonstrating oncogenic synergy in the growth of tumors carrying Ras mutations at codon 12 and codon 76 in Balb/c nude mice. Fig. 3C is expanded detail of the study in fig. 3B to show the growth of individual E76 mutant-transfected cells relative to BALB/3T3 cells transfected with wild-type *ras*. Bars are standard deviation of mean tumor volume from 3 mice per group.

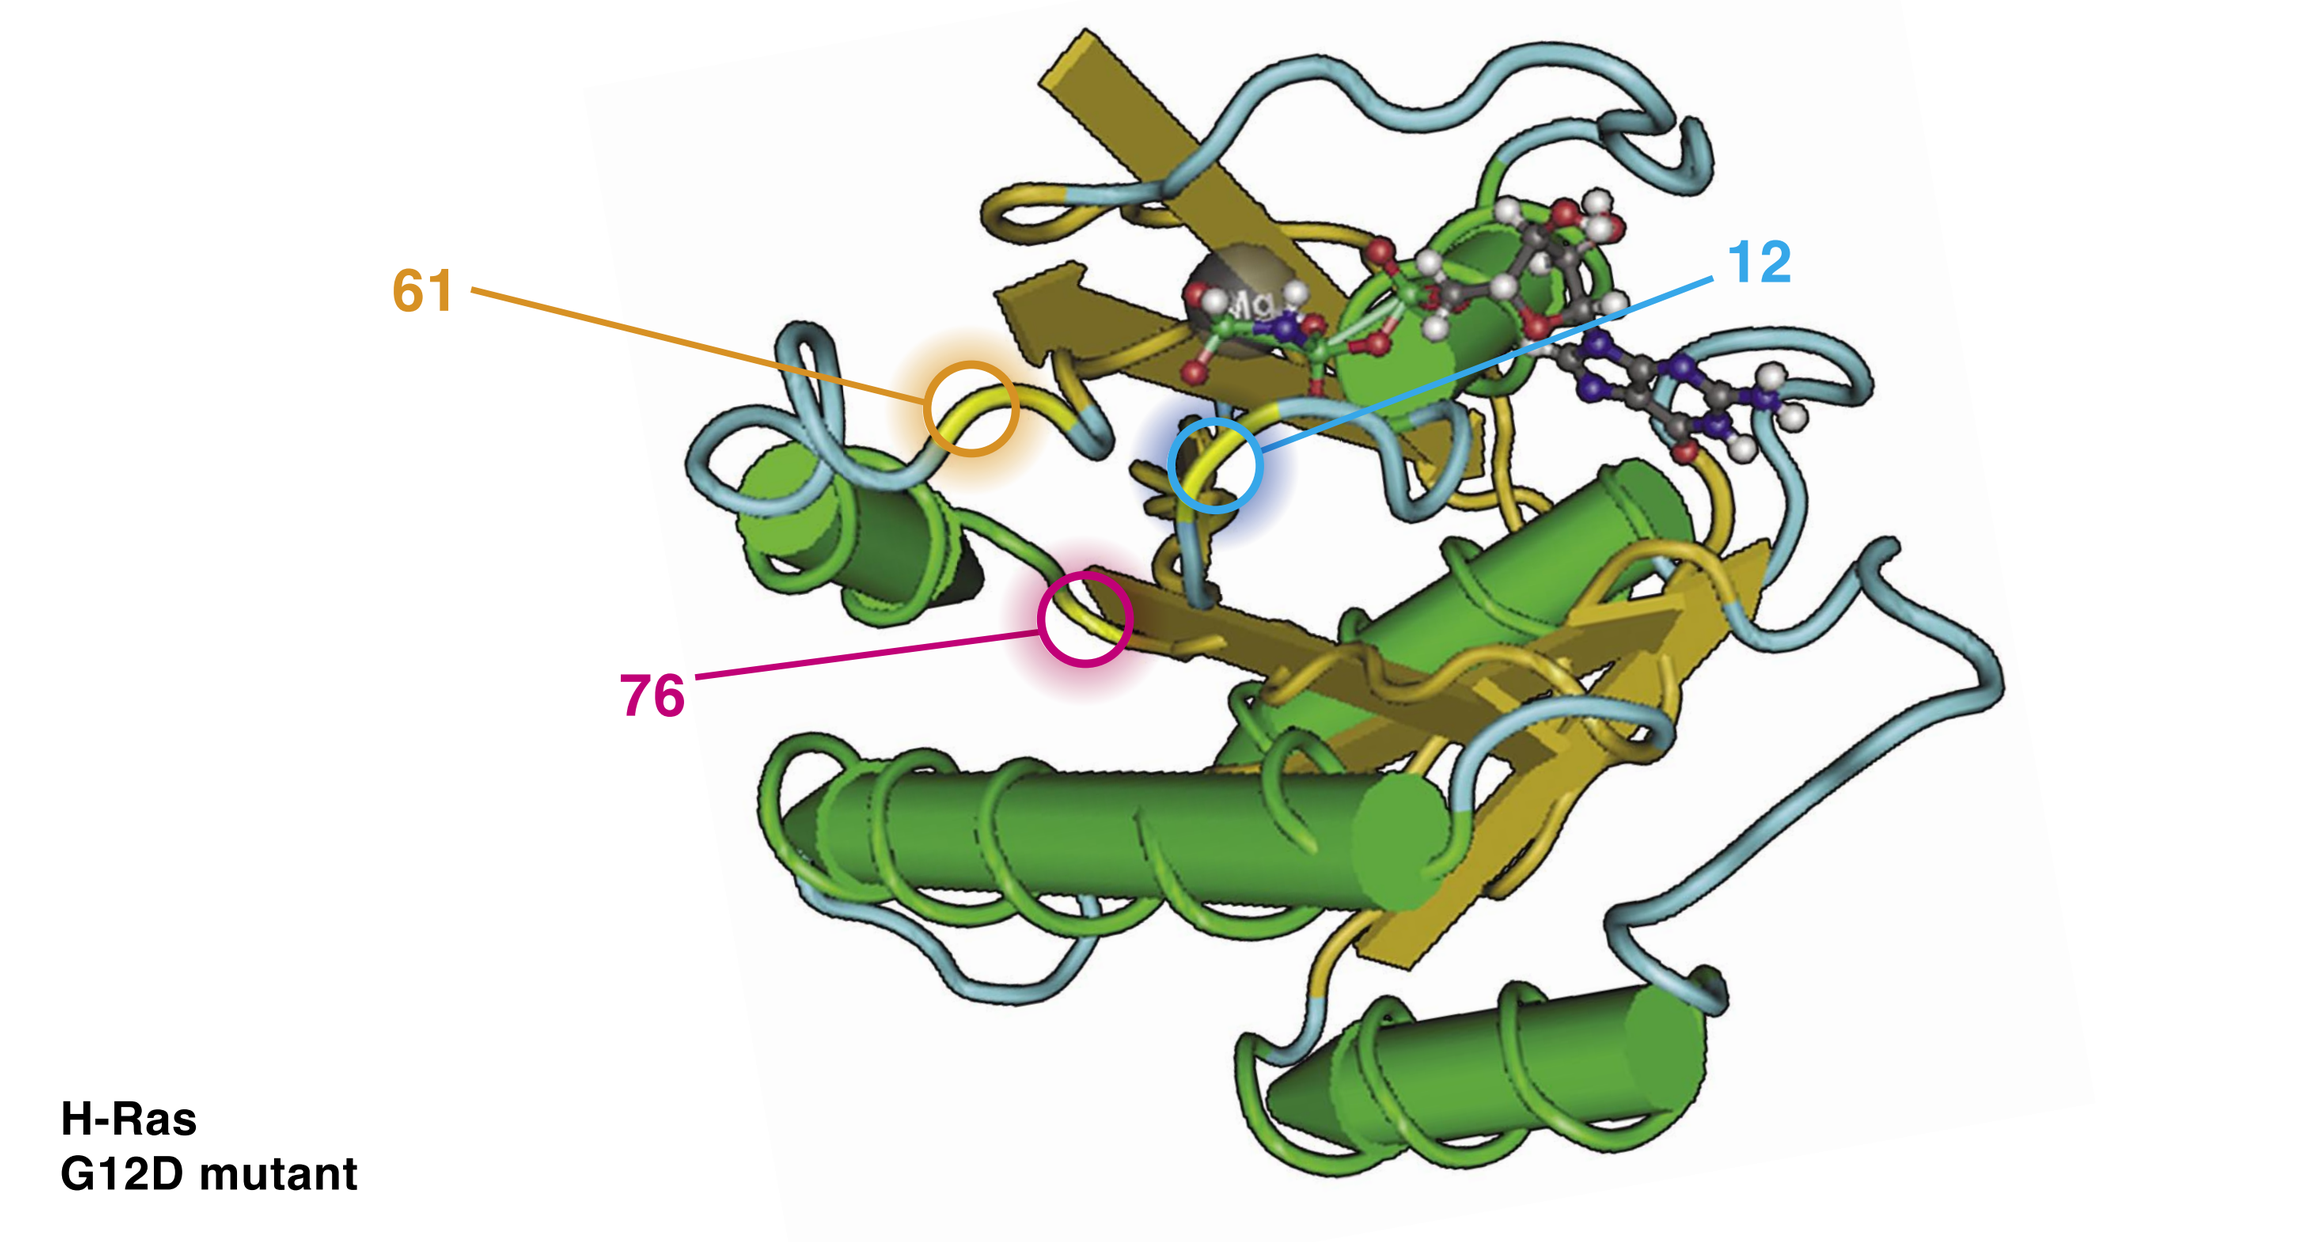
61
12
76
Mg
H-Ras
G12D mutant

Franken SM, Scheidig AJ, Krengel U, et al. Biochemistry v32, p.8411-8420.

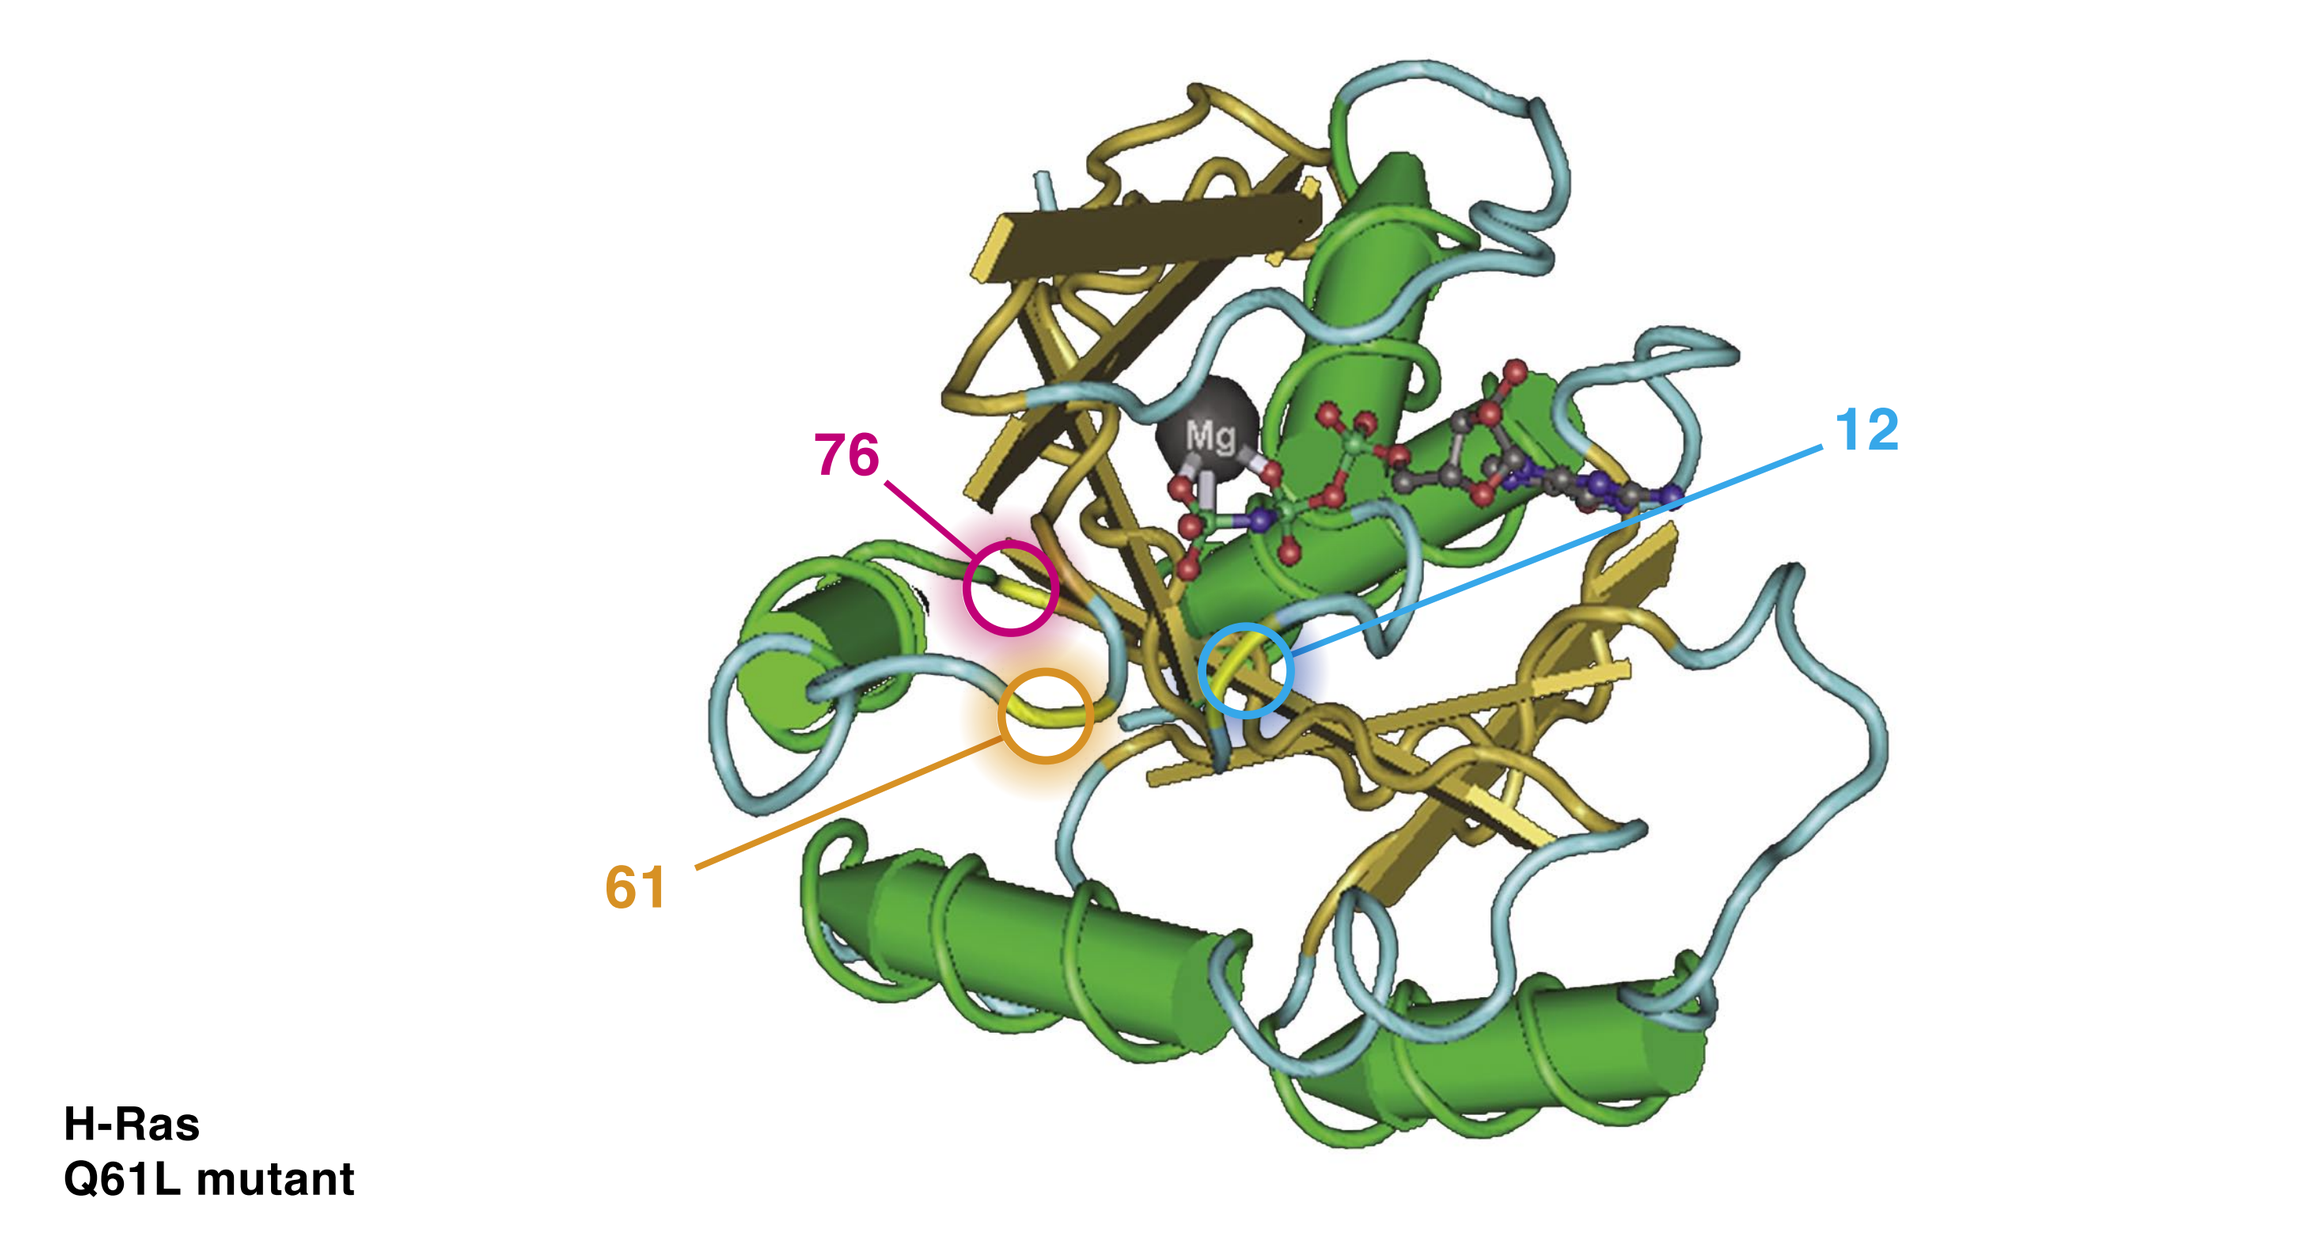
76
12
61
Mg
H-Ras
Q61L mutant

Krengel U, Schlichting L, Scherer A, et al. Cell v62, p.539-548.

Figure 4: Ras E76 mutations may affect L2 region conformation and Ras-GAP binding

This schematic drawing highlights the change in the Ras protein crystal structure due to a codon 12 mutation in H-Ras (G12D) (top) compared to the wild-type (G12) sequence for that domain (bottom) and codon 61 mutation (bottom) compared to the wild-type sequence for that domain (top). Amino acid 61 is located at the proximal end of the Switch 2/Loop4 domain, which is involved with binding to GTPase activating proteins (GAPs) that trigger the hydrolysis of Ras-GTP. The amino acid 76 position at the other 'hinge' of the critical Switch 2/Loop 4 (GAP-binding) domain, so mutations in this amino acid will similarly interfere with Ras function.

Conclusions

1. A new *ras* mutation at codon 76 was identified in 16 % tumors from pancreas, colorectal and lung cancer subjects in a phase 1 clinical trial. The amino acid at codon 76 was found mutated from glutamic acid to glycine, lysine or glutamine.
2. Ras E76 mutations cause cell transformation to a tumor phenotype *in vitro* and *in vivo.*
3. Ras E76 mutations synergize with Ras G12 mutations to exacerbate the aggressive tumor growth phenotype.
4. The presence of Ras E76 mutations may represent a diagnosis of accelerated malignancy.
5. Ras E76 mutations may be used as a new target antigen for cancer therapy.

©2007 GlobeImmune, Inc.