HCV-SPECIFIC CELLULAR IMMUNITY, RNA REDUCTIONS, AND NORMALIZATION OF ALT IN CHRONIC HCV SUBJECTS AFTER TREATMENT WITH GI-5005, A YEAST-BASED IMMUNOTHERAPY TARGETING NS3 AND CORE: A RANDOMIZED, DOUBLE-BLIND, PLACEBO CONTROLLED PHASE 1B STUDY

GlobeImmune
www.GlobeImmune.com

Schiff, Eugene R.[2]; Everson, Gregory T.[4]; Tsai, Naoky[8]; Bzowej, Nathalie H.[7]; Gish, Robert G.[7]; McHutchison, John[9]; Jacobson, Ira M.[5]; Tong, Myron J.[3]; Jensen, Donald M.[6]; Lauer, Georg M.[10]; Cruickshank, Scott[11]; Ferraro, John[1]; Haller, Aurelia[1]; Duke, Richard[1]; Rodell, Timothy[1]; Apelian, David[1];
Clinical Development and Regulatory Affairs, GlobeImmune, Louisville, CO, USA.[1] Center for Liver Diseases, University of Miami, Miami, FL, USA.[2] Huntington Medical Research Institutes, Pasadena, CA, USA.[3] University of Colorado School of Medicine, University of Colorado at Denver Health Sciences Center, Denver, CO, USA.[4] Weill Medical College of Cornell University1, New York, NY, USA.[5] University of Chicago, Chicago, IL, USA.[6] The Barry S. Levin, MD Department of Transplantation, California Pacific Medical Center, San Francisco, CA, USA.[7] Department of Medicine, University of Hawaii, Honolulu, HI, USA.[8] Duke Clinical Research Institute, Duke University, Durham, NC, USA.[9] Partners AIDS Research Center, Harvard Medical School, Boston, MA, USA.[10] Scott Cruickshank and Associates, Inc., Santa Barbara, CA, USA.[11]

Tarmogen Technology

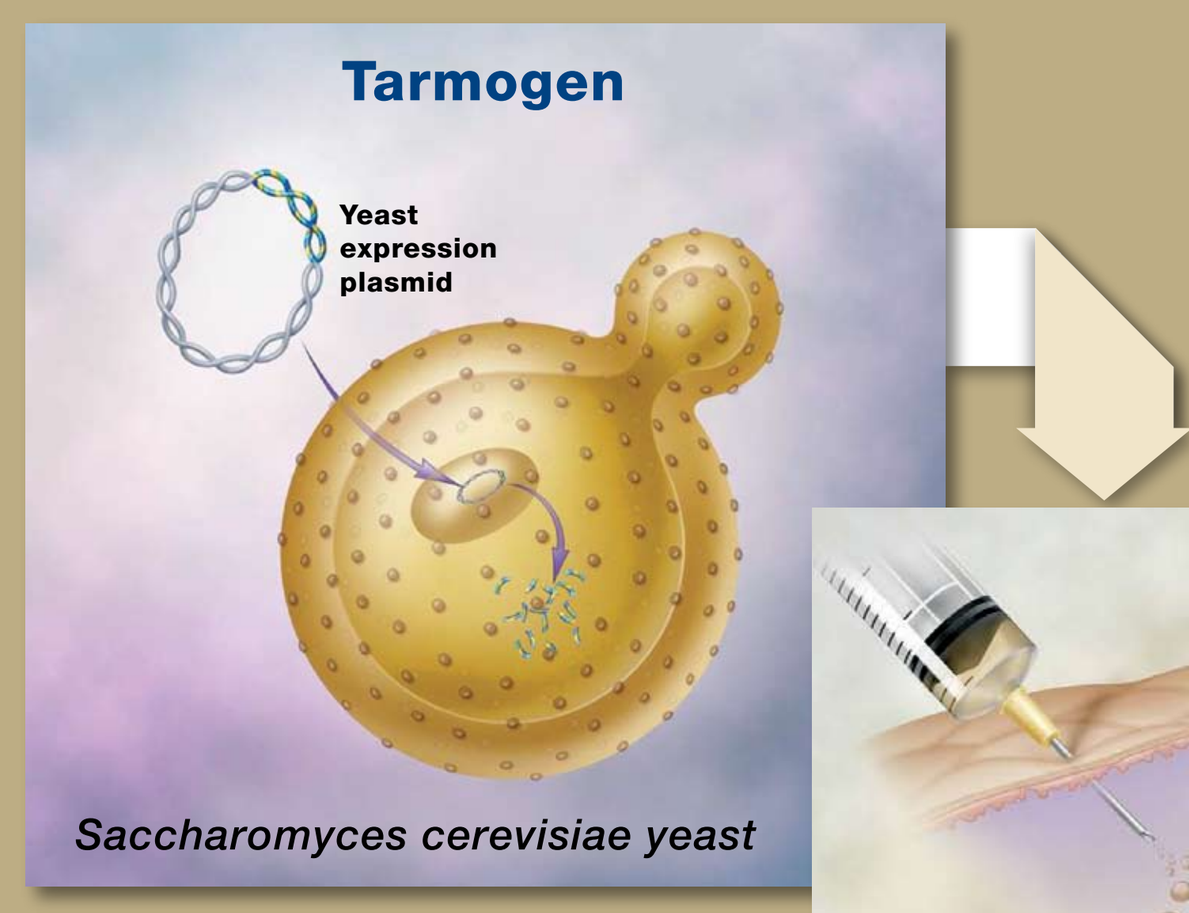
Tarmogen
Yeast expression plasmid
Saccharomyces cerevisiae yeast

Heat inactivated, recombinant *S. cerevisiae* expressing HCV NS3-Core protein antigens (GI-5005)

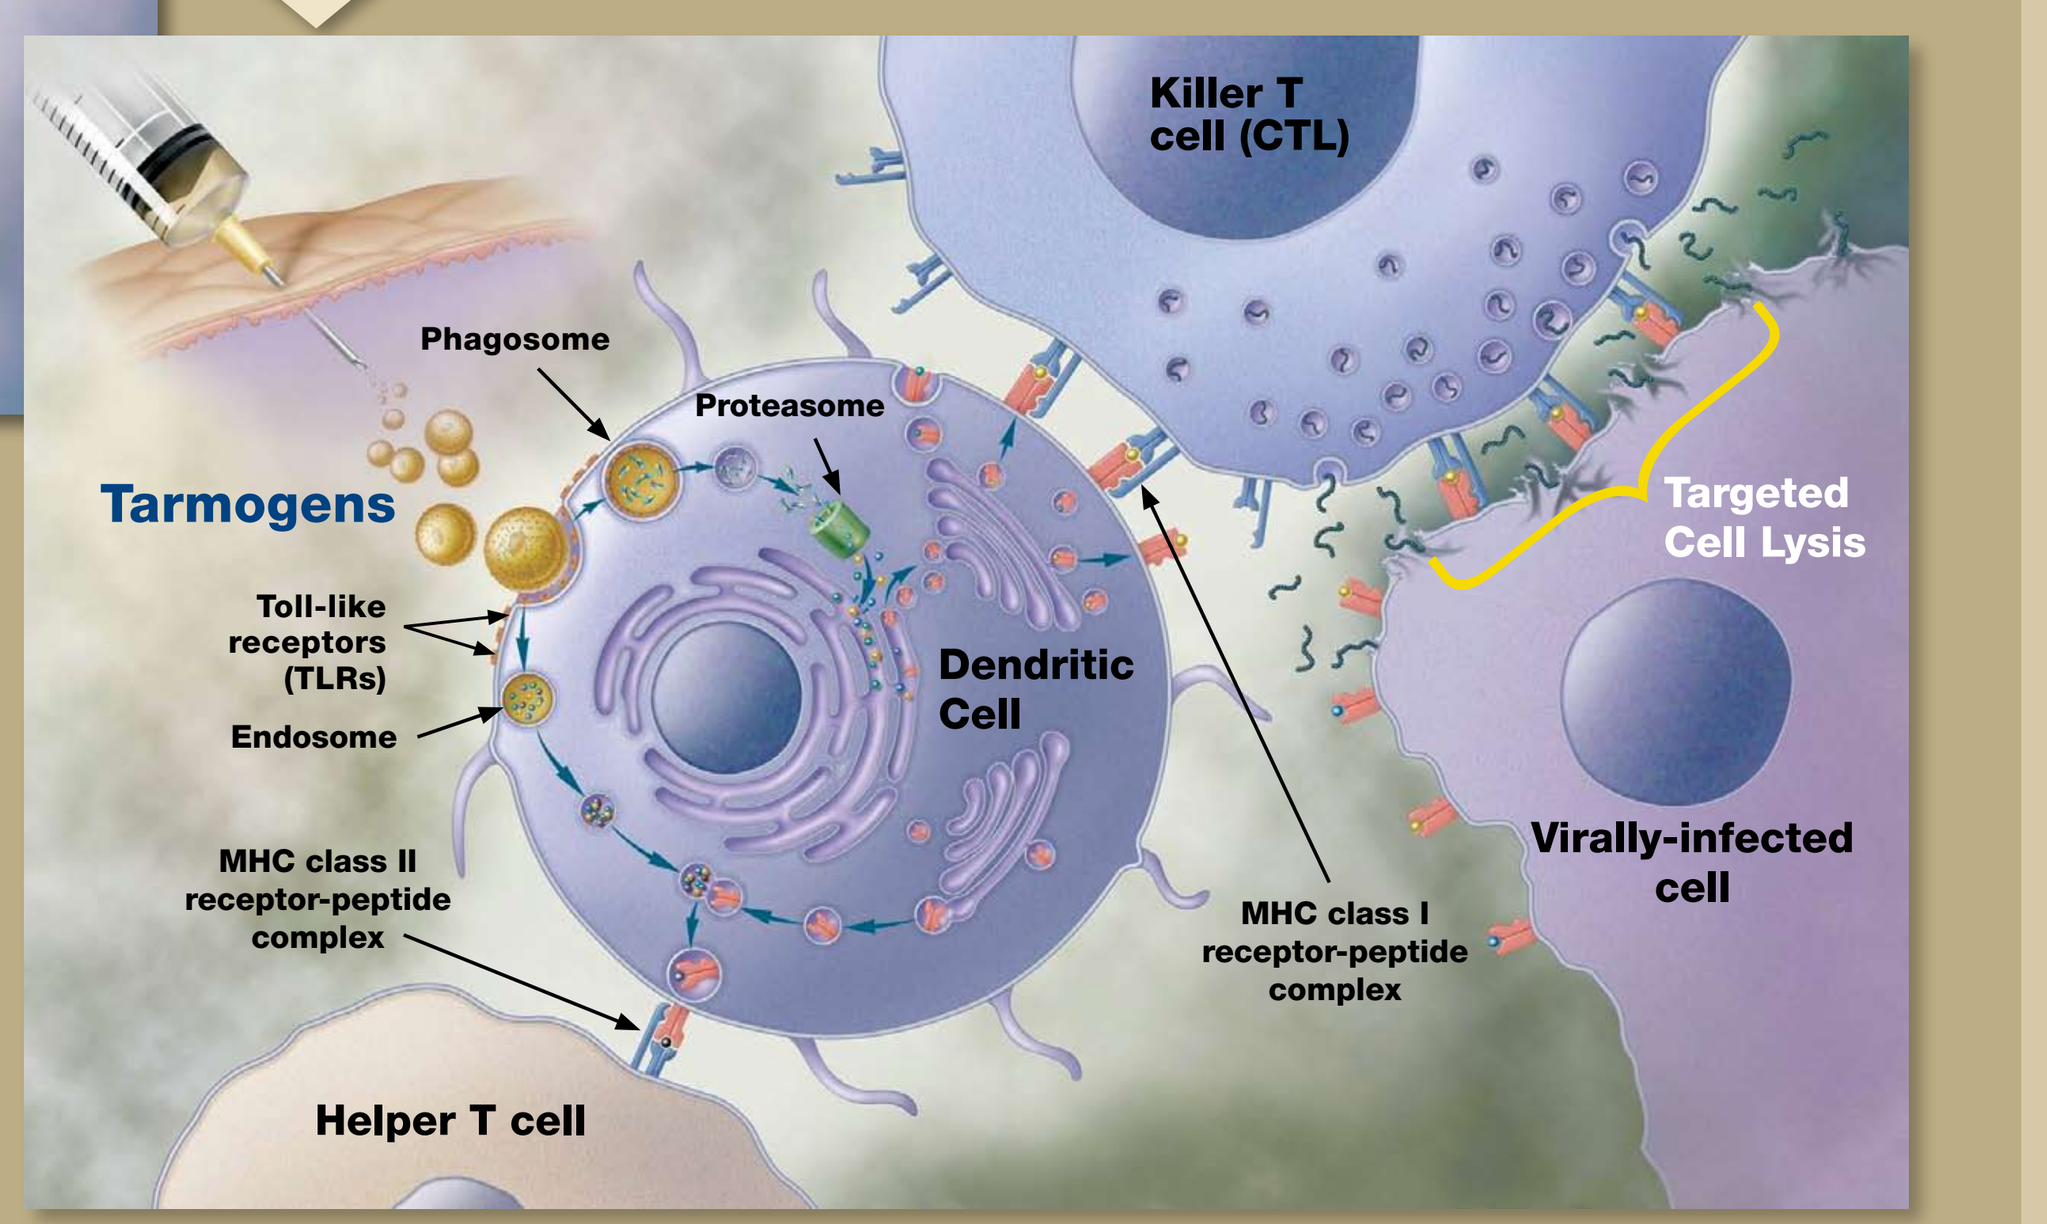
Killer T cell (CTL)
Phagosome
Proteasome
Tarmogens
Toll-like receptors (TLRs)
Endosome
Dendritic Cell
Targeted Cell Lysis
Virally-infected cell
MHC class II receptor-peptide complex
MHC class I receptor-peptide complex
Helper T cell

Uptake of Tarmogens by dendritic cells results in the activation of HCV antigen-specific helper and cytotoxic T cells.

Introduction

Tarmogens (targeted molecular immunogens) are whole, heat-killed recombinant *Saccharomyces cerevisiae* yeast engineered to express one or more target protein antigens, and activate both an innate immune response via Toll-like receptors (TLRs), as well as an adaptive, antigen-specific immune response. GI-5005 was engineered to express a fusion protein comprised of large segments of the hepatitis C virus (HCV) NS3 protease and Core proteins, including sequences which are essential and are highly conserved across the different HCV genotypes. GI-5005 was designed to induce a broad cellular immune response, which has been associated with HCV clearance in patients who spontaneously clear infection during the acute phase. The mechanism of action for GI-5005 (i.e. immune elimination of infected hepatic cells) may work synergistically in combination with the current or emerging standard of care which directly inhibits viral replication. Additionally, the GI-5005 mechanism of action may offer an option for interferon intolerant or contraindicated patients as a long term monotherapy.

In preclinical studies, GI-5005 has demonstrated:

1. Induction of potent HCV-specific cellular immune responses;
2. Dose-dependency of response, which is boosted with repeated immunizations;
3. No evidence of immune neutralization or immune-tolerance to the product;
4. Eradication of HCV-antigen expressing tumors in mice following prophylactic or therapeutic administration;
5. No significant toxicity with either GI-5005 or other Tarmogens in a number of species including mice, rats, rabbits and non-human primates

Study GI-5005-01 was designed to evaluate the subcutaneous administration of 7 doses of GI-5005, administered as monotherapy, in subjects with chronic hepatitis C infection who were either partial responders or relapsers to an interferon based regimen using either pegylated or non-pegylated interferon alpha with or without ribavirin, or treatment naïve. An interim report describing the results from 42 patients in the first four dose-cohorts (0.05-10YU) demonstrated robust HCV specific T cell responses in treated subjects as measured by ELISpot assay, a statistically significant difference in the ALT nadir of treated versus placebo patients, as well as several treated patients with viral load reductions between -0.75 and -1 log_{10} (Everson, Poster/Abstract #LB17 AASLD 2006). This report summarizes all of the GI-5005 dose groups including the highest doses tested (20YU and 40YU). The higher GI-5005 dose groups demonstrate a dose response for ALT normalization, higher amplitude ELISpot responses than the lower dose GI-5005 groups, and additional patients with viral load reductions in the -0.75 to -1.4 log_{10} range. These responses were not observed in any placebo patients.

Study Design

GI-5000-01 is a double-blind, placebo-controlled, multi-center, dose-escalation, therapeutic trial evaluating the subcutaneous administration of 7 doses of GI-5005 as a monotherapy in subjects with chronic hepatitis C infection who were either partial responders or relapsers to an interferon based regimen using either pegylated or non-pegylated interferon alpha with or without ribavirin, or treatment naïve. The initial dose of GI-5005 was 0.05YU (yeast units: 1YU = 10^7 yeast cells) delivered subcutaneously once per week for 5 consecutive weeks followed by two additional monthly doses. The subsequent dose groups were escalated to 0.5YU, 2.5YU, 10.0YU, 20YU and 40YU. Table 2 below summarizes the dose levels and planned group sizes.

Table 1

Treatment Period (GI-5005 monotherapy)	Monitoring/Follow up
1 8 15 22 29 36 43 57 64 71 85	92 99 169 225 336
• Viral load and ALT testing on all visits	
• Immunology (ELISPOT and LPA) at baseline and days 36, 99, 225	
• 71 subjects were enrolled in 6 dose groups at 9 centers in the U.S.	
Dosing Days (1, 8, 15, 22, 29, 57, 85); Monitoring (36, 43, 64, 71, 92, 99, 169, 225, 336)	

Table 2

Dose Group	Number of subjects (active:placebo)	GI-5005 Yeast units (YU)
Group 1	6:2	0.05
Group 2	6:2	0.5
Group 3	6:2	2.5
Group 4	12:4	10.0
Group 5	12:4	20.0
Group 6	12:4	40.0

* 1YU = 1 yeast unit =10^7 yeast cells

Demographics

Table 3

Variable	Placebo	GI-5005 Dose Level (YU)						
		0.05	0.5	2.5	10.0	20.0	40.0	Combined
Number of subjects	19	5	6	6	12	11	12	52
Age (Mean)	54.0	47.0	51.0	53.5	53.0	57.0	52.5	54.0
Gender (F / M)	8 / 11	1 / 4	2 / 4	3 / 3	3 / 9	4 / 7	5 / 7	18 / 34
HCV Genotype (1 / 2 / 3)*	19 / 0 / 0	5 / 0 / 0	6 / 0 / 0	5 / 1 / 0	10 / 2 / 0	8 / 2 / 1	11 / 1 / 0	45 / 6 /1
Prior HCV treatment response								
Relapsed	9 (47.4%)	2	6	4	6	7	7	32 (61.5%)
Partial responder	7 (36.8%)	2	0	0	2	2	4	10 (19.2%)
Naïve	2 (10.5%)	1	0	2	4	1	1	9 (17.3%)
Null responder	1 (5.3%)	0	0	0	0	1	0	1 (1.9%)
Pre-study liver histology								
Evidence of inflammation (Y / N)	18 / 1	4 / 1	6 / 0	6 / 0	12 / 0	11 / 0	10 / 2	49 / 3
Evidence of fibrosis (Y / N / ND)	16 / 3 / 0	5 / 0 / 0	6 / 0 / 0	6 / 0 / 0	11 / 1 / 0	11 / 0 / 0	7 / 4 / 1	46 / 5 / 1
Evidence of cirrhosis (Y / N)	0 / 19	0 / 5	0 / 6	0 / 6	0 / 12	0 / 11	0 / 12	0 / 52
Median Baseline HCV RNA (x 1000 IU/ml)	2,440	2,290	921	766	853	3,010	2,210	1,140
Median Baseline ALT (U/L)	57.0	68.0	71.5	94.5	78.0	49.0	51.0	61.0

[a] genotype 1 includes 1, 1a and 1b; genotype 2 includes 2a and 2b

Safety Summary

GI-5005 was well tolerated, and the dose escalation was completed to the highest planned dose without dose limiting toxicity. Safety findings were generally limited to transient local injection site reactions and transient mild constitutional complaints.

HCV Specific Immune Response

Using stringent criteria for grading HCV specific immune response by ELISpot assay we have observed responses only in the GI-5005 treated patients. The strongest amplitude responses against an individual peptide pools (>250 activated cells per million lymphocytes) were observed only in the highest GI-5005 dose groups (10YU, 20YU, and 40YU). See Table 4 for details.

ELISpot Criteria

Non-optimized peptide ELISpot:
- 15 or more pools ≥ 25 spots
- Or at least 10 pools ≥ 25 spots with at least 2 pools positive on more than 1 on-treatment measurement
- Or at least 5 pools ≥ 25 spots with at least one pool ≥ 150 spots

Optimized peptide ELISpot:
- 4 or more pools ≥ 75 spots
- Or at least 2 pools ≥ 75 spots and at least 1 pool positive on more than 1 on-treatment measurement
- Or at least 2 pools ≥ 75 spots and at least 1 pool ≥ 150 spots

A treatment-emergent HCV specific immune response was observed in GI-5005 treated subjects, but not placebo subjects using ELISpot assay. ELISpot assay, a measure of T lymphocyte activation, was run on peripheral blood mononuclear cells (PBMCs) from subjects at select study visits (Day 1, 36, 92, and 225). The PBMCs were mixed with HCV peptides *ex vivo* and analyzed for interferon γ (IFNγ) production, a hallmark of T cell activation. Pools of non-optimized HCV peptides (15-mers to 20-mers) as well as pools of optimized HCV peptides (8 to 10-mers) which represent all of the genes of the hepatitis C genome were used for *ex vivo* stimulation of subject PBMCs. PBMCs were then harvested after ex vivo peptide stimulation and the number of cells (or "spots") per million PBMCs that produce IFNγ was measured using enzyme linked immunosorbant assay.

To assess for the amplitude, breadth, and persistence of responses across the ELISpot pools, the peptide pools of individual subjects were assessed by correcting each Day 36, 92, and 225 peptide pool score by subtracting the baseline value for that given peptide pool. Nine of 39 treated subjects (23%) with sufficient immune sampling met the following definition of responder see (above) for the non-optimized peptide conditions and/or the optimized peptide conditions. None of the 16 placebo subjects met these criteria for ELISpot response.

Table 4

ELISpot Conditions	Placebo	0.05 YU	0.5 YU	2.5 YU	10 YU	20 YU	40 YU	All treated
Non-optimized or optimized	0/16	1/5	2/6	0/6	4/10	1/7	1/5	9/39 (23%)
>250 cells/million	0/16	0/5	0/6	0/6	2/10 (20%)	2/7 (29%)	1/5 (20%)	5/39 (13%)
>150 cells/million	2/16 (12.5%)	1/5 (20%)	2/6 (33%)	2/6 (33%)	5/10 (50%)	4/7 (57%)	1/5 (20%)	15/39 (38%)

*The 40YU group is less mature and has fewer follow-up measurements for ELISpot assay resulting in a potential under-reporting of responses by ELISpot

Viral Load Data

To date in this study, six GI-5005 treated subjects* had reductions in HCV RNA from baseline (-0.75 log_{10} to -1.4 log_{10}). No subjects treated with placebo had HCV RNA reductions ≥ 0.75 log_{10}.

Figure 5

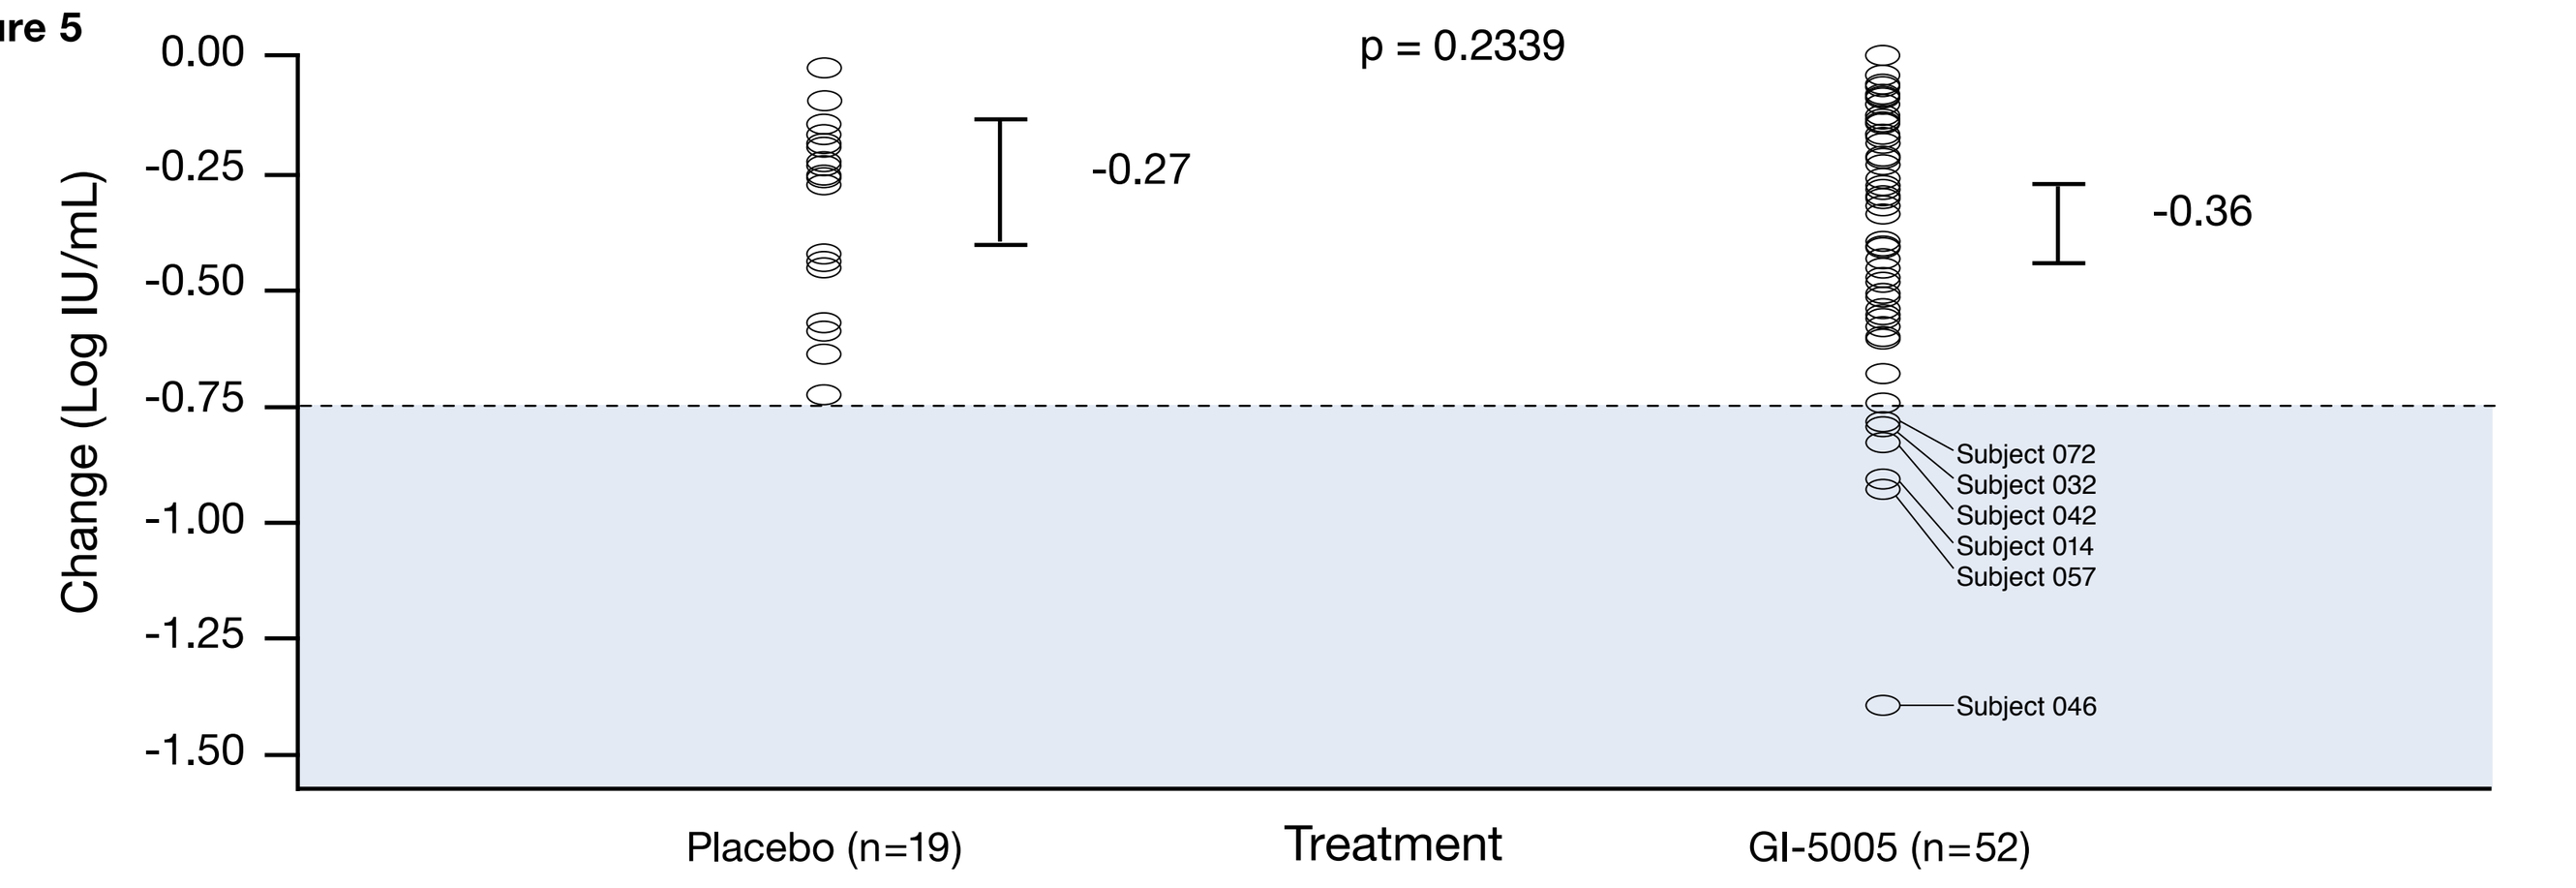
p = 0.2339
0.00
-0.25
-0.50
-0.75
-1.00
-1.25
-1.50
Change (Log IU/mL)
-0.27
-0.36
Subject 072
Subject 032
Subject 042
Subject 014
Subject 057
Subject 046
Placebo (n=19)
Treatment
GI-5005 (n=52)

*Four subjects were genotype 1, one subject was genotype 2, and one subject was genotype 3 which is approximately the ratio of genotypes enrolled in the GI-5005 treatment group

ALT Normalization

Alanine aminotransferase (ALT) is an enzyme found in high quantities in the liver. It is a well validated measure of hepatic injury and serves as a surrogate for hepatic inflammation. Reductions and/or normalization of ALT levels have been shown to correlate with improved hepatic inflammation and fibrosis as determined by serial biopsy. In this study we have observed a dose response for ALT normalization, with normalization defined as at least 2 consecutive visits with ALT within normal limits in those patients with baseline ALT greater than the upper limit of normal.

Figure 6

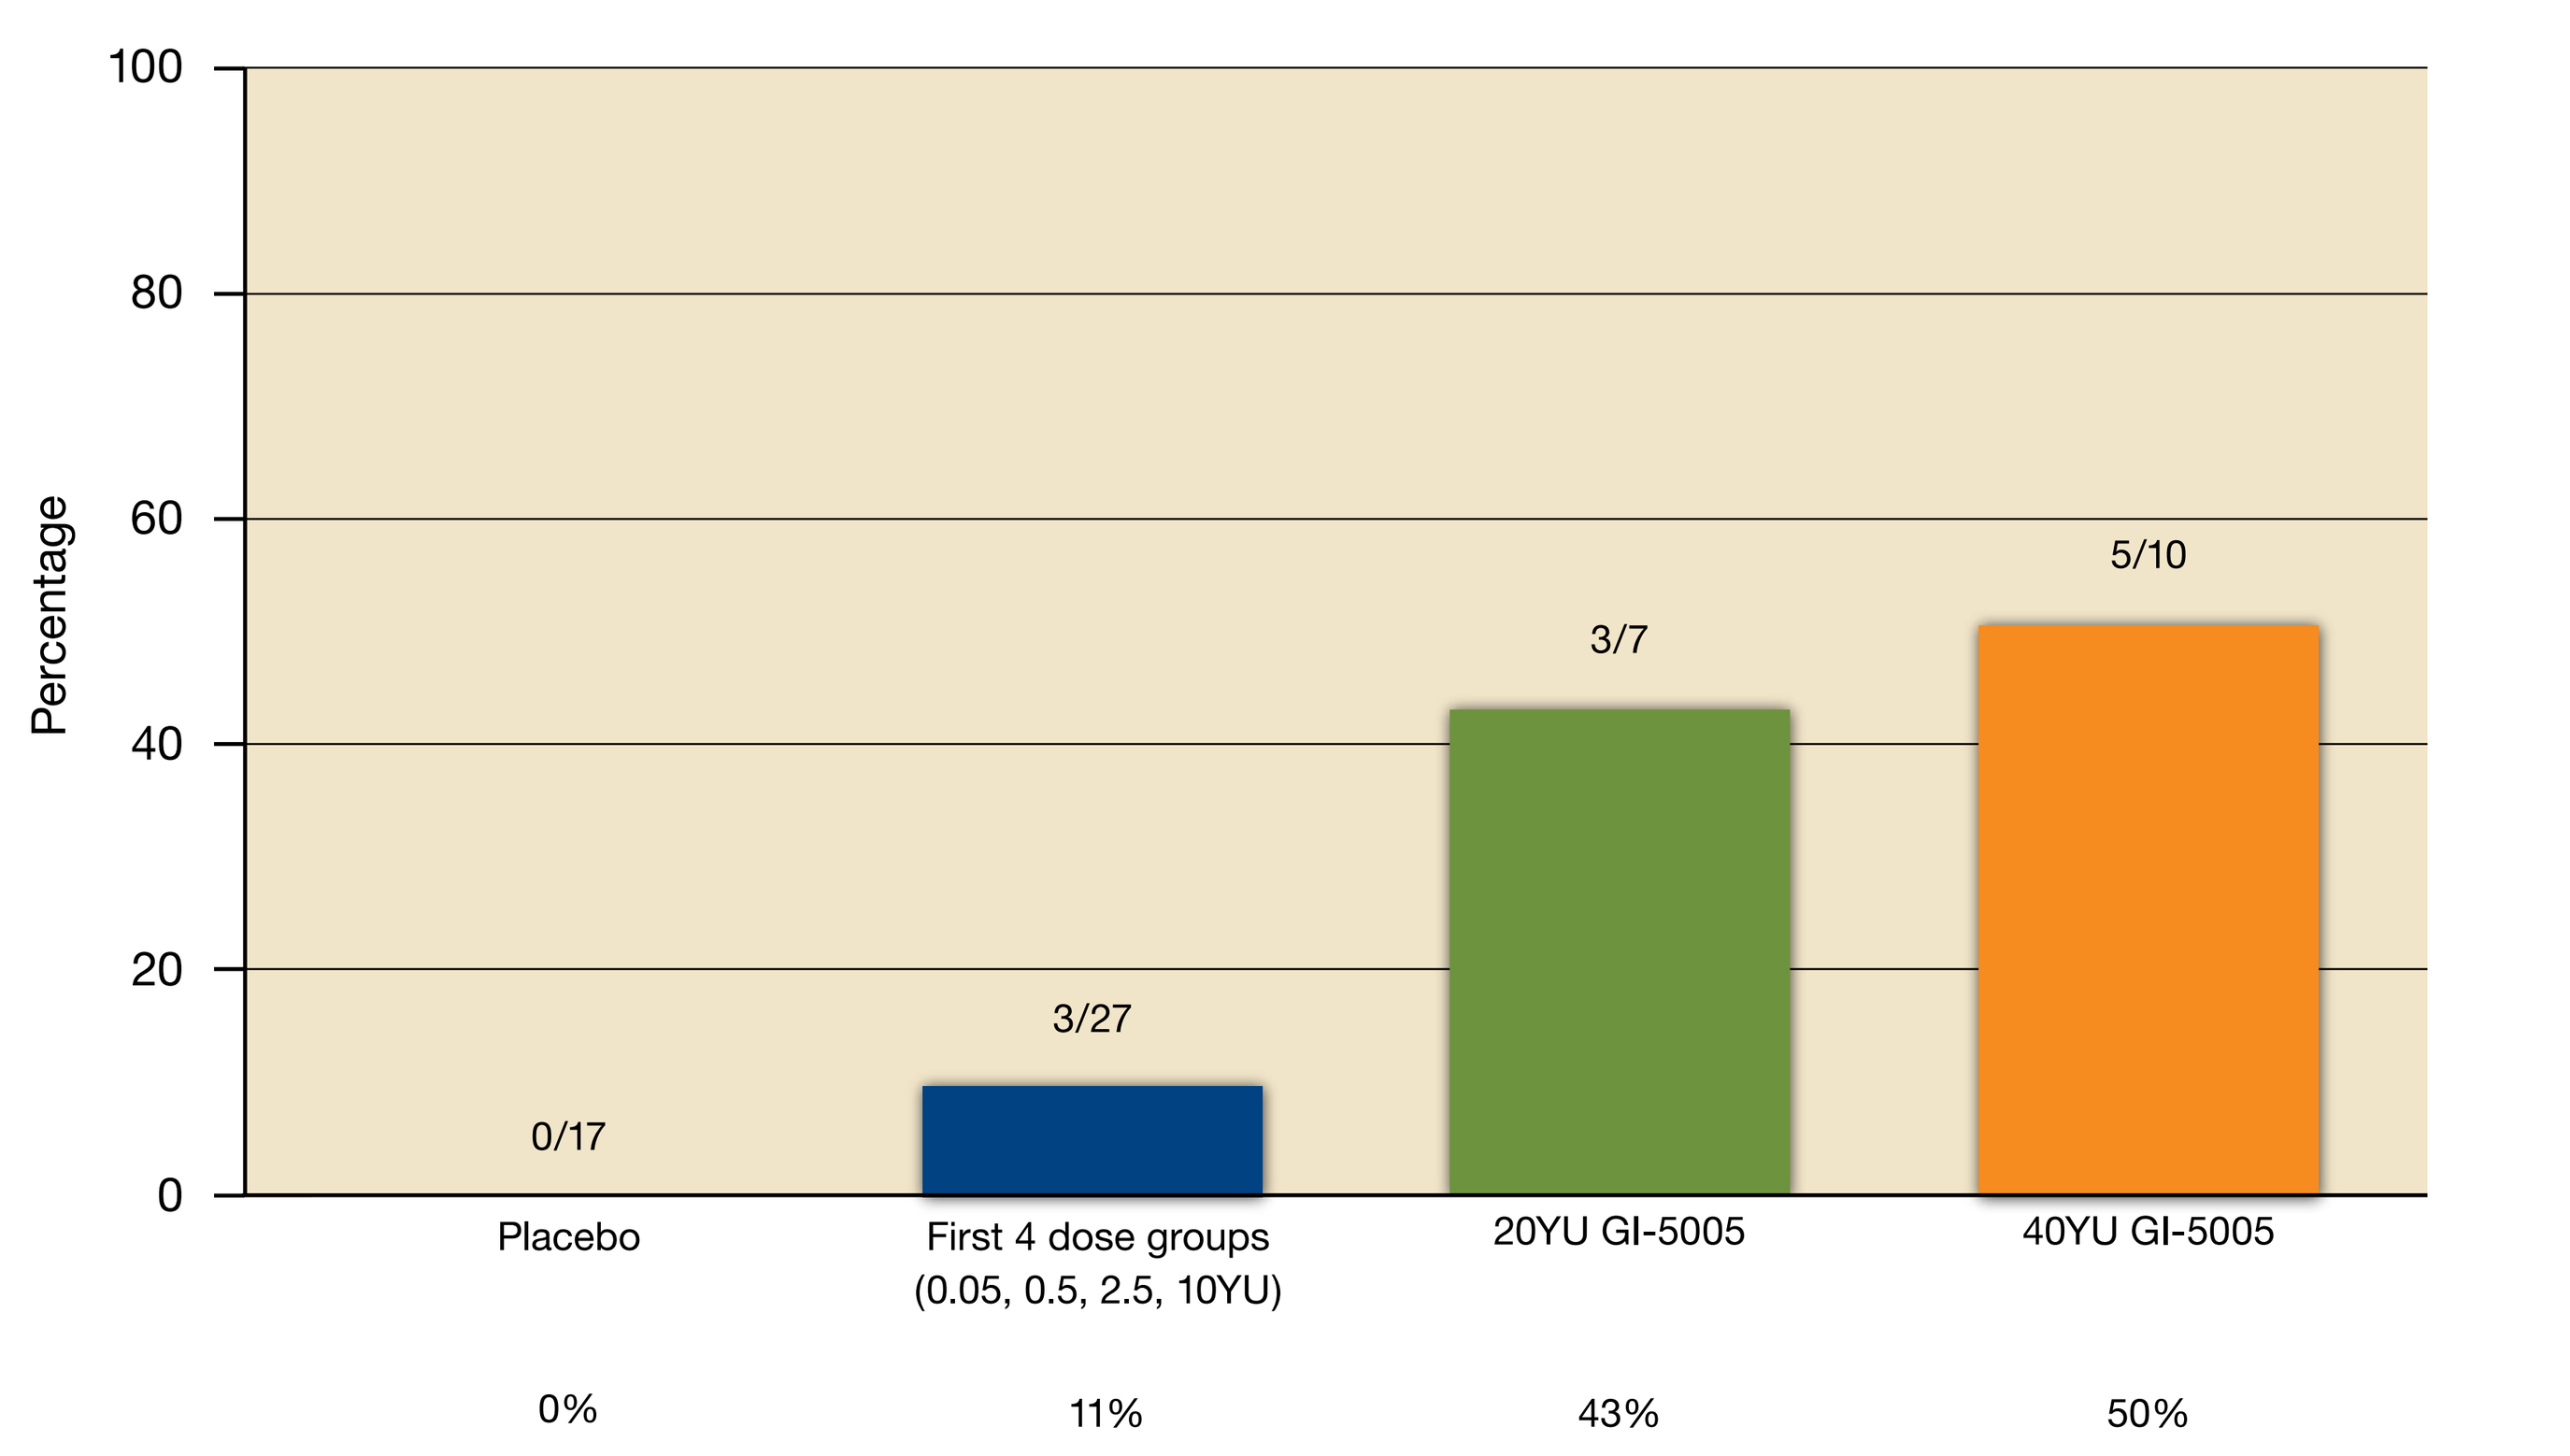
100
80
60
40
20
0
Percentage
0/17
3/27
3/7
5/10
Placebo
First 4 dose groups (0.05, 0.5, 2.5, 10YU)
20YU GI-5005
40YU GI-5005
0%
11%
43%
50%

Conclusion

Safety

- GI-5005 was well tolerated, with no dose limiting toxicities (DLTs) through 40YU.

Efficacy

- Viral load reductions from -0.75 to -1.4 log_{10} were observed only in GI-5005 treated patients 6/54 (11%)
- GI-5005 dose response for ALT normalization reaching 50% in the 40YU group. No ALT normalization has been observed in the placebo group.
- HCV specific cellular immune responses only observed in GI-5005 treated subjects (9/39, 23%) using stringent criteria for amplitude and breadth of immune response. The strongest ELISpot responses (> 250 activated cells per million lymphocytes) were detected only in the highest GI-5005 doses tested (10YU, 20YU, and 40YU)

These results indicate that a short course of GI-5005 monotherapy is capable of generating an HCV specific immune response that is associated with viral load reductions of up to 1.4 log_{10}, and ALT normalization in up to half of the high dose patients. A Phase 2 trial comparing GI-5005 plus pegylated interferon/ribavrin versus pegylated interferon/ribavirin alone is being initiated at 50 centers in the US, EU, and India. Long-term GI-5005 salvage will also be provided in this trial to patients who fail to achieve an early virologic response (EVR) or do not tolerate treatment in the pegylated interferon/ribavirin arm.

GlobeImmune

GlobeImmune, Inc
1450 Infinite Drive
Louisville, Colorado 80027
tel 303-625-2700
fax 303-625-2710
www.globeimmune.com

Abstract

Purpose: Evaluation of the efficacy, immunogenicity, and safety of GI-5005 in subjects with chronic HCV infection.

Methods: GI-5005 is a whole heat-inactivated *S. cerevisiae* immunotherapy expressing HCV NS3 and Core. Subjects with chronic HCV infection who were interferon (IFN) partial responders, relapsers, or treatment naïve were eligible. Five weekly subcutaneous (SC) doses of GI-5005 monotherapy over 29 days were followed by two monthly SC GI-5005 doses and 9 months of post-treatment follow-up. Dose groups of 0.05, 0.5, 2.5, 10, 20, and 40YU (1 YU = 10,000,000 yeast cells) were randomized 3:1 treated:placebo.

Results: Dose escalation was completed without a product-related dose limiting toxicity, serious adverse event, or exacerbation of hepatitis (66 subjects total; 6 in 0.05YU, 6 in 0.5YU, 7 in 2.5YU, 12 in 10.0YU, 11 in 20YU, 7 in 40YU, and 17 in placebo). HCV-specific cellular immune responses with broad epitope coverage were observed by ELISpot assay in 33% of treated subjects (8/24) to date with a dose response for GI-5005, compared to no responses in the placebo group. Statistically significant improvements in ALT levels were observed in treated subjects with a mean maximum decrease from baseline of 29.3% versus 16.9% for placebo (47 treated versus 17 placebo respectively, p=0.02). Three of the 7 (43%) 20YU patients with abnormal ALT at baseline had normalized their ALT after treatment (>2 consecutive visits WNL) compared to 4 of 29 treated subjects (14%) from the first 4 dose groups combined, and none of the placebo patients. Six subjects of 47 treated overall (13%) achieved a viral load nadir ranging from -0.75 to -1.04 $\log_{10}$ change from baseline with the 2 greatest reductions (2/11 subjects, 18%) observed in the 20YU group (-0.93 $\log_{10}$ and -1.04 $\log_{10}$). In several cases these viral load reductions persisted for months and in one case for the full 9 month post-treatment follow-up period. No HCV RNA reductions in -0.75 $\log_{10}$ to -1 $\log_{10}$ range were observed in the placebo group.

Addendum: Additional data were collected in the GI-5005 20YU and 40YU dose groups which further support the GI-5005-associated trends for HCV antigen-specific cellular immune response, HVC RNA reductions, and ALT normalization. While these data were collected and included in the analysis after submission of the above abstract, they have been included in the poster presentation.

Conclusion: GI-5005 was well tolerated and generated HCV-specific cellular immune responses with treatment-associated HCV RNA reductions and ALT normalizations and dose-dependent normalization of ALT in a subset of patients, which were not observed in the placebo group. These data support further development of GI-5005 in combination with IFN-based standard of care, or as a salvage treatment in patients who are intolerant or who fail IFN-based regimens. Future evaluation of combination of GI-5005 with small molecule inhibitors of HCV replication is also warranted based on its complementary immune mechanism of action.