Recombinant yeast (*Saccharomyces cerevisiae*) vector encoding carcinoembryonic antigen (CEA) activates human dendritic cells for CEA-specific T-cell responses

Cinzia Remondo[1], Vittore Cereda[1], Alex Franzusoff[2], Jeffrey Schlom[1] and Kwong Y. Tsang[1]

[1]Laboratory of Tumor Immunology and Biology, Center for Cancer Research, National Cancer Institute, NIH, [2]GlobeImmune, Inc., 145 Infinite Dr., Louisville, CO 80027

BACKGROUND

Various strategies have been studied to enhance DCs ability to activate antigen specific T cells. It has been demonstrated that antigens can be engineered for heterologous expression in ***Saccharomyces cerevisiae*** yeast by cloning the gene encoding the target antigen into a yeast-expression plasmid, under the control of an inducible or constitutive promoter. In addition, it has been demonstrated that whole recombinant yeast vaccine can activate human DCs and induce phenotypic maturation of human DCs derived from monocytes, increasing surface expression of CD80, CD86, CD40, CD54, and MHC class II, and produce inflammatory cytokines such as IL-12 and TNF-α, stimulating strong alloreactive T-cell proliferation. This study analyzes the ability of a *S. cerevisiae* yeast vector containing a transgene encoding carcinoembryonic antigen (yeast-CEA) to activate human DCs and stimulate antigen-specific T-cell responses.

RESULTS

QUESTION 1: How does yeast-CEA affect the maturation of human dendritic cells?

1. Treatment of immature human DCs with yeast-CEA results in upregulation of CD80, CD83, CD54, CD58, class I and class II.

Treatment of DCs	CD80	CD83	CD54	CD58	Class I	Class II
UNTREATED	18.1% (57.7)	38.0% (85.6)	88.3% (115.6)	86.4% (98.1)	98.9% (627.4)	82.3% (558.4)
CD40L	80.0% (47.8)	86.5% (51.8)	99.9% (434.7)	99.9% (278.7)	99.8% (1,403.8)	99.9% (434.7)
CONTROL YEAST	83.2% (36.8)	81.4% (51.7)	99.9% (155.0)	99.3% (106.0)	99.9% (1,232.7)	92.7% (370.0)
YEAST-CEA	74.3% (25.5)	77.0% (42.5)	99.9% (144.5)	98.9% (94.1)	99.5% (1,214.4)	96.4% (270.9)

Flow cytometric analysis of DCs for surface markers expression Human DCs were cultured for 48 h with media only (untreated), stimulated with CD40L, treated with control yeast or yeast-CEA. DCs were harvested and analyzed for surface-marker expression; results are shown in %-positive cells and MFI (parentheses).

2. Treatment of immature human DCs with yeast-CEA results in enhanced production of cytokines and chemokines.

Treatment of DCs	IL-12p70	TNF-α	IFN-γ	IL-8	IL-2	IL-4	IL-13	IL-5	IL-10	IL-1β
UNTREATED	0	1	0	44	2	177	11	0	0	0
CD40L	693	8,969	735	526	136	51	233	24	113	21
CONTROL YEAST	3,503	>10,000	5,349	5,827	375	70	72	4	85	49
YEAST-CEA	5,914	>10,000	6,718	7,148	773	42	131	6	141	79

Human DCs were cultured with media only (untreated), stimulated with CD40L, or treated with control yeast or yeast-CEA. Cultured supernatants were collected and screened for cytokine and chemokine production by multiplex cytokine/chemokine kit; results are expressed in pg/mL.

CONCLUSION 1

Treatment of immature human dendritic cells with yeast-CEA results in enhanced expression of CD80, CD83, CD54 and CD58 as well as enhanced production of IL-12p70, TNF-α, IFN-γ, IL-1β, IL-8 and IL-2.

QUESTION 2: Can yeast-CEA and yeast-CEA(6D)[CEA gene with an agonist epitope]-treated dendritic cells activate CEA-specific T cells?

1. Yeast-CEA-treated DCs can activate CEA-specific T cells to produce high levels of IFN-γ. Yeast-CEA(6D)-treated DCs can activate CEA-specific T cells to produce higher levels of IFN-γ compared to DCs treated with yeast-CEA.

A.

DCs	DCs treatment	CEA(6D) peptide	CEA-specific T cells (V8T)	IFN-γ (pg/ml)
–	–	–	+	<15.6
+	–	–	–	<15.6
+	–	–	+	<15.6
+	–	+	+	47.4
+	CD40L	–	–	<15.6
+	CD40L	–	+	<15.6
+	CD40L	+	+	535.5
+	Control yeast	–	–	<15.6
+	Control yeast	–	+	<15.6
+	Control yeast	+	+	534.3

B.

DCs	DCs treatment	CEA-specific T cells (T-VLG)	IFN-γ (pg/ml)
–	–	+	<15.6
+	Yeast-CEA	–	<15.6
+	Yeast-CEA(6D)	–	<15.6
+	Yeast-CEA	+	508.7
+	Yeast-CEA(6D)	+	1,501.8

A. DCs treated with CD40L were cultured for 24 h, then pulsed with CEA-6D peptide. DCs treated with control yeast were pulsed with CEA-6D peptide. CEA-specific V8T cells were stimulated with peptide-pulsed or unpulsed DCs. *B.* DCs were treated with yeast-CEA or yeast-CEA(6D) and then used to stimulate a CEA-specific T-VLG cell line. Supernatants were screened for production of IFN-γ by ELISA. Yeast-CEA(6D) is a recombinant *S. cerevisiae* construct expressing CEA-6D, a HLA-A2-binding CEA agonist peptide.

2. CEA-specific CTLs can be established using DCs treated with yeast-CEA as antigen-presenting cells. The CEA-specific CTLs can kill tumor cells expressing CEA in a HLA-A2-restricted manner.

A.

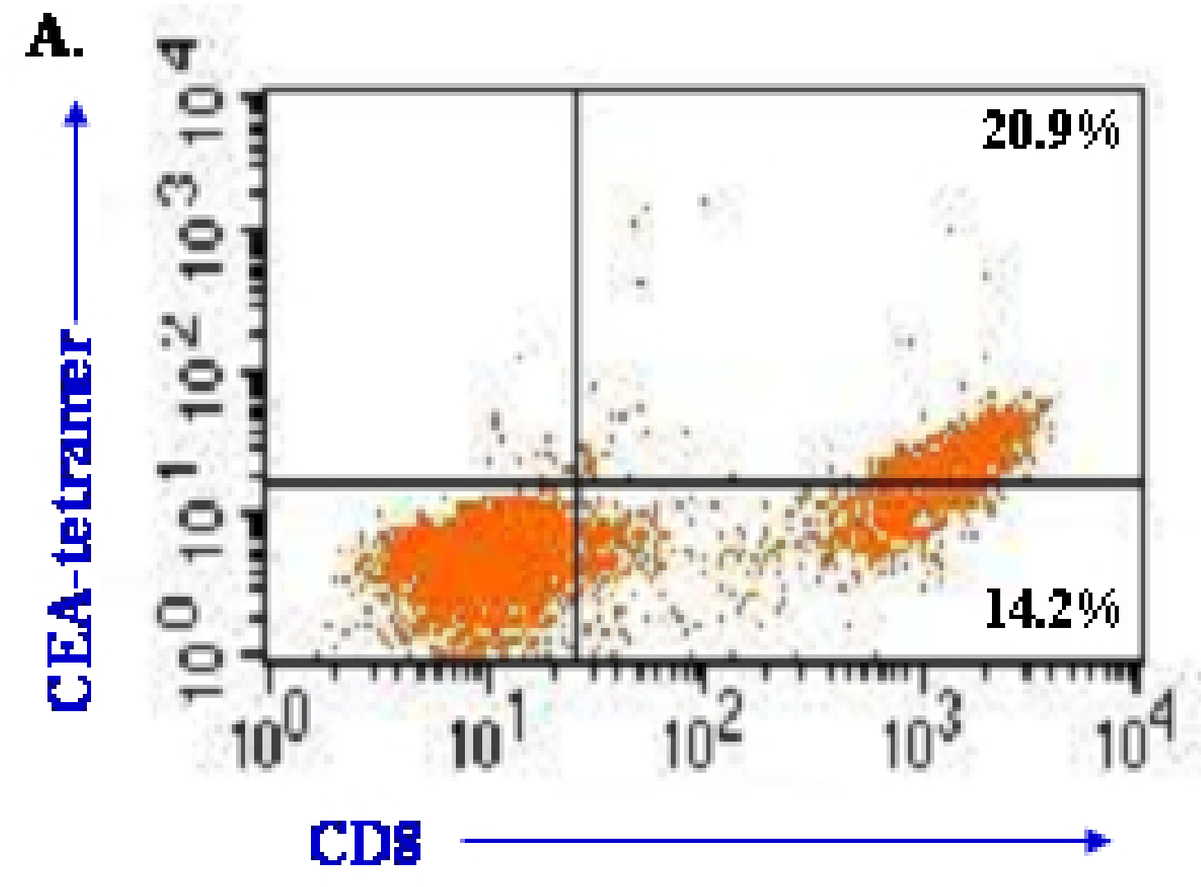
20.9%
14.2%
CEA-tetramer
CD8

B.

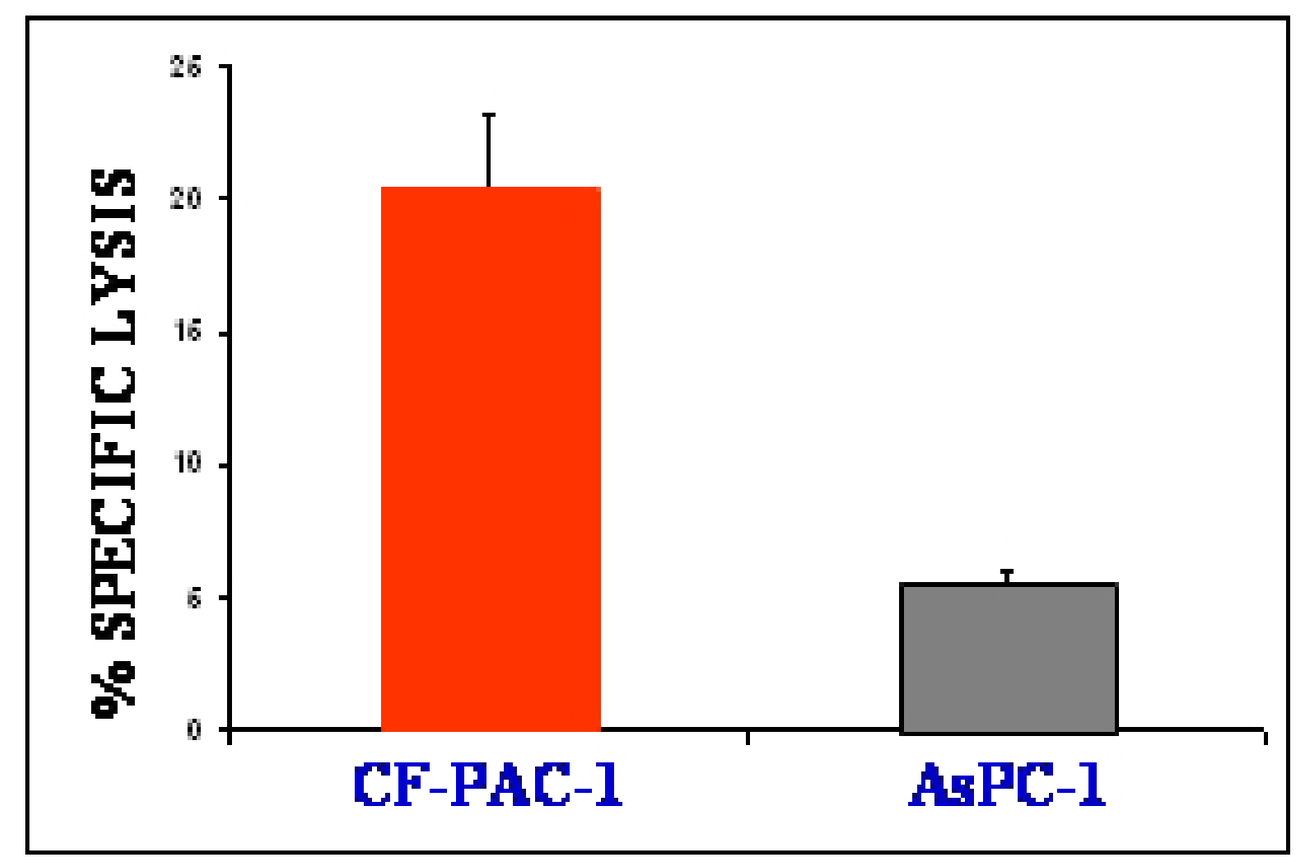
% SPECIFIC LYSIS
CF-PAC-1
AsPC-1

DCs obtained from a patient with breast carcinoma were treated with yeast-CEA. *A.* After IVS-3, cells were analyzed by flow cytometry for their ability to bind CEA-tetramer. *B.* A 16-h 111Indium release assay was performed using CD8+/CEA-tetramer+ T cells, with CF-PAC1 (HLA-A2+, CEA+) and AsPC-1 (HLA-A2–, CEA+) tumor cell lines as targets. Results are expressed as percentage of specific lysis at an E:T ratio of 25:1.

CONCLUSION 2

Human DCs treated with yeast-CEA can activate CEA-specific CTLs. Yeast-CEA(6D) can activate CEA-specific T cells to produce higher levels of IFN-γ compared to CEA-specific T cells activated by DCs treated with yeast-CEA.

Human DCs treated with yeast-CEA can function as APCs to generate CEA-specific T-cell lines capable of lysing CEA$^+$ human tumor cells.

QUESTION 3: What is the gene expression profile of DCs treated with yeast-CEA?

1. Gene expression profile of DCs treated with yeast-CEA at 8 hours and 24 hours.

Functional genes groups		8 hours	24 hours
CHEMOKINES AND THEIR RECEPTORS	*CCL2*	8.5	41.9
	CCL20	4.8	8.1
	CCL3L1	/	6.9
	CCL4	/	32.3
	CCL5	12.9	77.1
	CCL8	19.3	51.9
	CCR7	/	12.9
	CXCL1	6.9	/
	CXCL10	59.6	276.6
	CXCL11	51.5	431.3
	CXCL2	/	3.5
	CXCL9	4.7	6.5
	IL8	/	17.2
CYTOKINES	*EBI3*	/	3.2
	IFNB1	7.9	/
	IL12A	/	4.3
	IL12B	162.8	65.5
	IL23A	/	8.5
	IL27	13.5	/
	IL29	3.5	/
	IL6	47.1	10.5
	TNF	29.9	17.3

Functional genes groups		8 hours	24 hours
ANTIGEN UPTAKE	*MARCKS*	4.0	10.8
	PNRC1	/	4.9
	SOD2	/	10.9
	TAP1	8.3	/
ANTIGEN PRESENTATION	*CD80*	/	3.3
	LAMP3	5.0	5.8
CELL SURFACE RECEPTORS	*CD40*	3.7	/
	LY75	/	5.1
SIGNAL TRANSDUCTION	*BTG1*	/	5.0
	GBP2	/	3.9
	GBP3	/	3.5
	IFI6	8.4	5.8
	IFI16	/	3.5
	IFI35	1.7	5.0
	IFI44	6	5.6
	IFIT1	9.8	22.5
	IFIT2	4.0	/
	IFIT3	7.8	2.9
	ISG15	8.6	10.3
	ISG20	28.7	96.7
	LTA	5.2	/
	NFKB1	/	5.2

Human DCs were generated from PBMCs of healthy donors and cultured with yeast-CEA at a DC:yeast-CEA. RNA was isolated and analyzed by Oligo GEArray® for human DC and APC microarray expressing 260 genes. Results represent ≥ 3-fold increases compared to untreated DCs. / = no change.

CONCLUSION 3

Treatment with yeast-CEA activates human DCs, as indicated by the upregulation of genes involved in the production of chemokines and cytokines and their receptors, as well as genes related to antigens uptake, antigen presentation and signal transduction.

SUMMARY

Heat-killed recombinant ***Saccharomyces cerevisiae*** vector expressing CEA (yeast-CEA) can activate human DCs inducing phenotypic maturation; it can also increase expression of genes involved in the production of chemokines and cytokines and their receptors, as well as genes related to antigen uptake, antigen presentation, and signal transduction. To our knowledge, ours is the first study to analyze the gene expression of DCs treated with yeast-CEA. Human DCs treated with yeast-CEA can activate CEA-specific CTLs that are capable of lysing tumor cells expressing CEA on a HLA-A2 restricted manner.